Phase 1 study of CB-839, a first-in-class, orally administered small molecule inhibitor of glutaminase in patients with refractory solid tumors

Meric-Bernstam F[1], DeMichele A[2], Telli ML[3], Munster P[4], Orford KW[5], Demetri GD[6], Schwartz GK[7], Iliopoulos O[8], Mier JW[9], Owonikoko TK[10], McKay RR[6], Mittra ES[3], Bennett MK[5], Patel MR[11,12], Infante JR[12], and Harding JJ[13]

[1]MD Anderson Cancer Center, Houston, TX; [2]Univ. of Pennsylvania, Philadelphia, PA; [3]Stanford Univ. Med. Center, Palo Alto, CA; [4]Univ. California, San Francisco, CA; [5]Calithera Biosciences, South San Francisco, CA; [6]Dana Farber Cancer Inst., Boston, MA; [7]Columbia Univ. Med. Center, New York, NY; [8]Massachusetts General Hospital, Boston, MA; [9]Beth Israel Deaconess Med. Center, Boston, MA; [10]Emory Univ. School of Medicine, Atlanta, GA; [11]Florida Cancer Specialists, Sarasota, FL; [12]Sarah Cannon Research Inst., Nashville, TN; [13]Memorial Sloan Kettering Cancer Center, New York, NY

BACKGROUND AND RATIONALE

- Glutamine is required for the growth and survival of many tumor types[1,2]
- Glutaminase (GLS) controls the first step in glutamine utilization in cancer cells (Figure 1), leading to the formation of glutamate, which is used to:
 - generate TCA cycle intermediates
 - synthesize glutathione
 - generate NADPH and maintain redox balance
 - synthesize anabolic building blocks, including nucleotides and fatty acids
- CB-839 is a highly selective, reversible, allosteric inhibitor of GLS[3]
- CB-839 has broad preclinical *in vitro* and *in vivo* anti-tumor activity in solid and hematologic malignancies[3,4]
- Herein we describe the initial results from CX-839-001, a first-in-man Phase 1 study of CB-839 in advanced solid tumors (ClinicalTrials.gov Identifier: NCT02071862)

Figure 1: Altered Glucose and Glutamine Metabolism of Cancer Cells

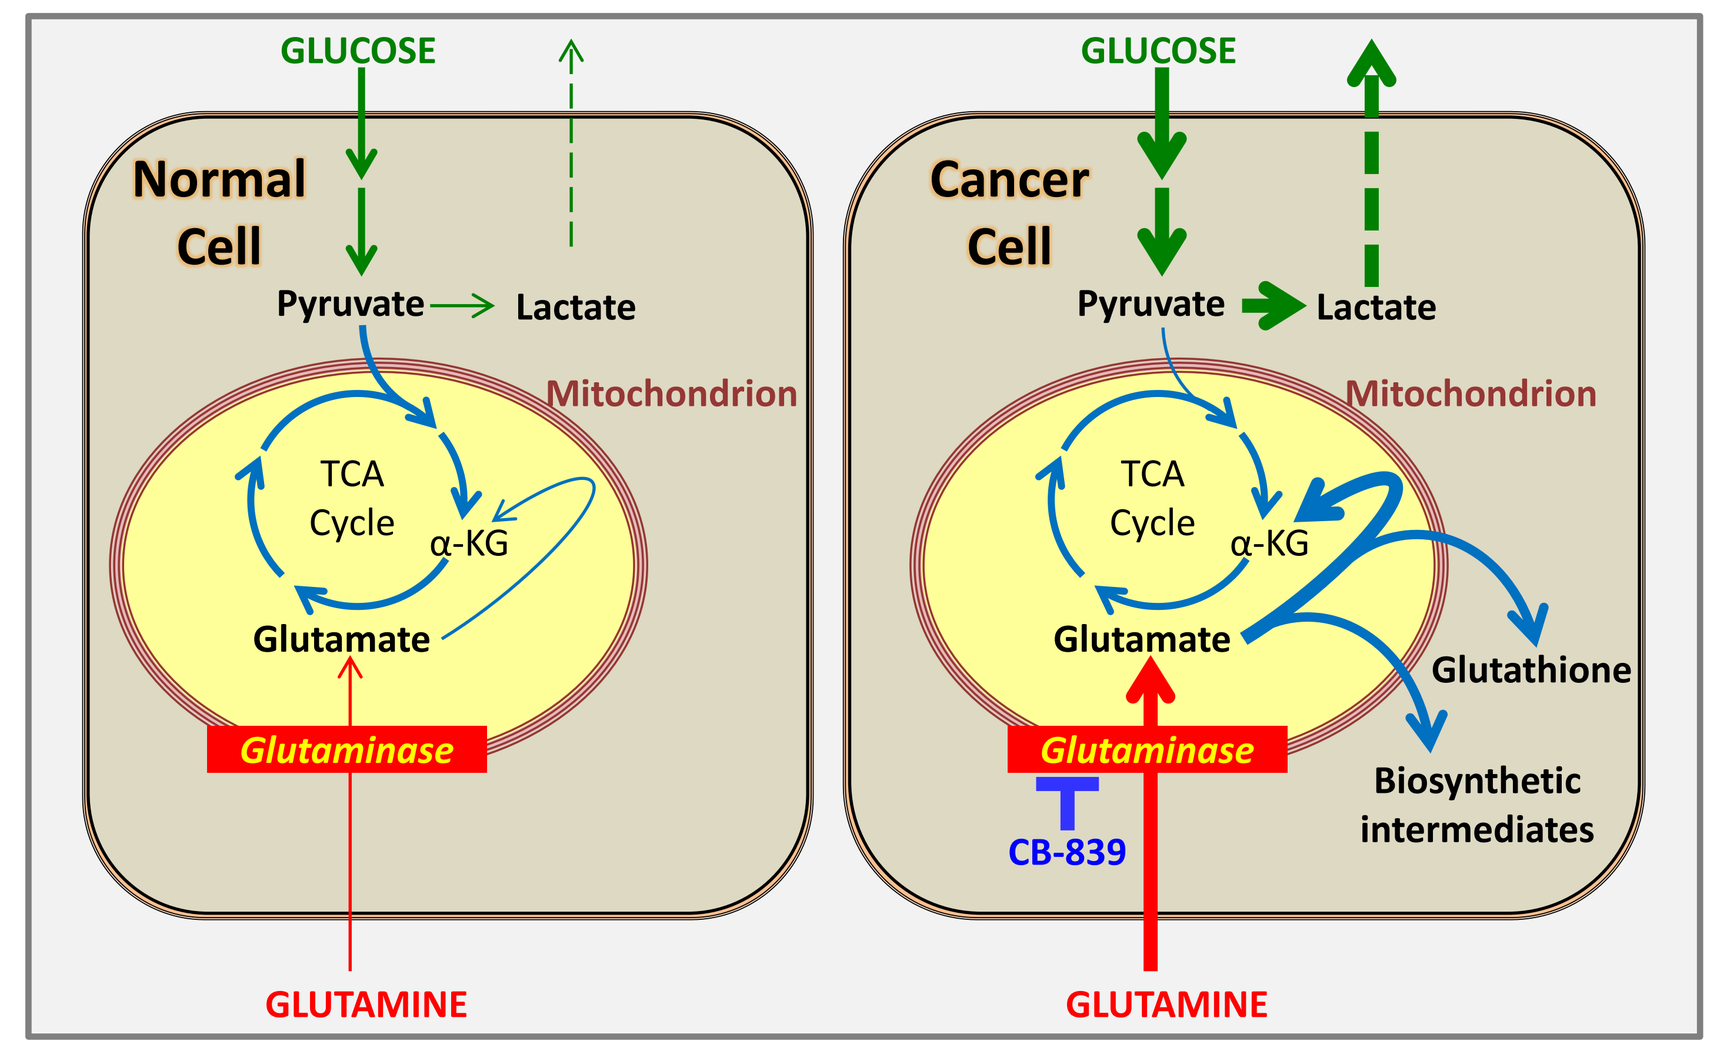
GLUCOSE
Normal Cell
Pyruvate
Lactate
Mitochondrion
TCA Cycle
α-KG
Glutamate
Glutaminase
GLUTAMINE
GLUCOSE
Cancer Cell
Pyruvate
Lactate
Mitochondrion
TCA Cycle
α-KG
Glutamate
Glutathione
Glutaminase
CB-839
Biosynthetic intermediates
GLUTAMINE

STUDY OBJECTIVES

Primary

- To evaluate the safety and tolerability and determine the single-agent Recommended Phase 2 Dose (RP2D) of CB-839 for locally-advanced, metastatic and/or refractory solid tumors

Secondary

- To determine the pharmacokinetics (PK) of CB-839 as a single agent
- To evaluate anti-tumor activity of the single-agent in patients with solid tumors

METHODS

Study Design

- Phase 1 study of CB-839 as a single-agent and in combination* with standard therapies in advanced solid tumors
 - Standard 3+3 dose escalation design with 3 week cycle length
 - Disease assessments are performed every three (3) cycles
- Expansion Cohorts for monotherapy in defined patient populations (see Figure 2)
 - Objectives include confirmation of safety profile and evaluation of clinical efficacy in defined patient populations
 - Simon two-stage design utilized for each expansion cohort
 - Planned enrollment of 11 patients per cohort
 - Further expansion with one response

** Only single-agent data presented in this poster*

Figure 2: CX-839-001 Study Design

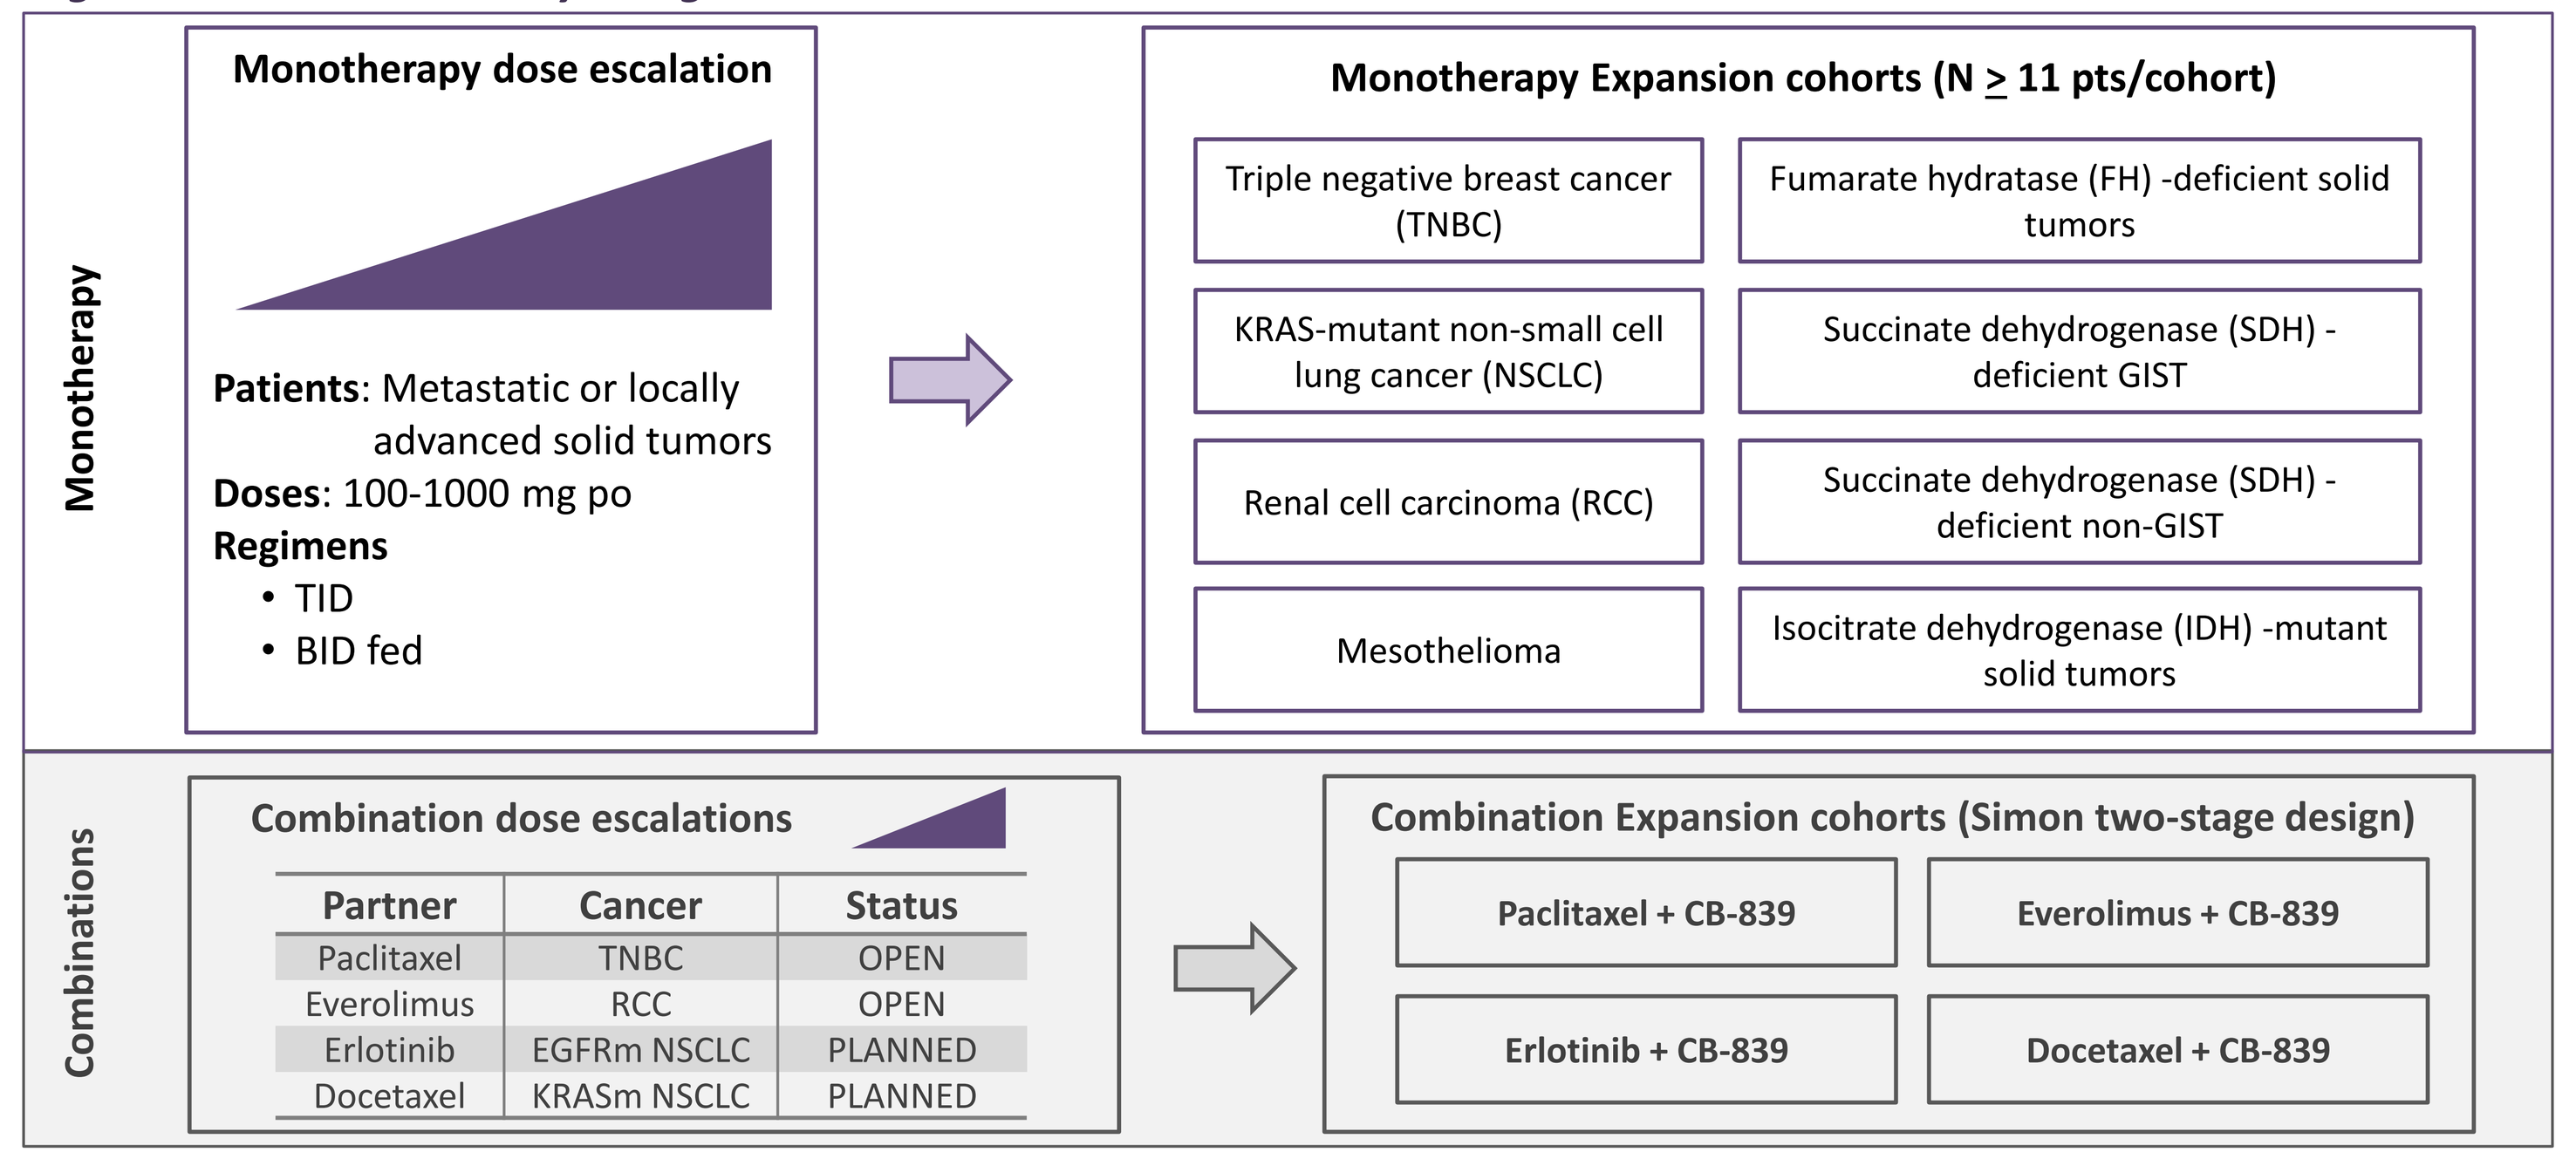
Monotherapy
Monotherapy dose escalation
Patients: Metastatic or locally advanced solid tumors
Doses: 100-1000 mg po
Regimens
• TID
• BID fed
Monotherapy Expansion cohorts (N ≥ 11 pts/cohort)
Triple negative breast cancer (TNBC)
Fumarate hydratase (FH) -deficient solid tumors
KRAS-mutant non-small cell lung cancer (NSCLC)
Succinate dehydrogenase (SDH) - deficient GIST
Renal cell carcinoma (RCC)
Succinate dehydrogenase (SDH) - deficient non-GIST
Mesothelioma
Isocitrate dehydrogenase (IDH) -mutant solid tumors
Combinations
Combination dose escalations
Partner | Cancer | Status
Paclitaxel | TNBC | OPEN
Everolimus | RCC | OPEN
Erlotinib | EGFRm NSCLC | PLANNED
Docetaxel | KRASm NSCLC | PLANNED
Combination Expansion cohorts (Simon two-stage design)
Paclitaxel + CB-839
Everolimus + CB-839
Erlotinib + CB-839
Docetaxel + CB-839

Key Inclusion Criteria

- Eastern Cooperative Oncology Group (ECOG) Performance Status of 0-1
- Life expectancy of at least 3 months
- Adequate hepatic, renal and hematological function
- Measurable Disease per RECISTv1.1

Key Exclusion Criteria

- Prior therapies within the specified time frames (14-21 days, depending on the agent)
- Any conditions that may preclude adequate absorption of study drug
- Untreated or unstable brain metastases

Study Treatment

- Oral CB-839 administered in 21-day cycles using one of two regimens:
 - **TID**: Three times daily (upon waking, at ~3 pm and at bedtime)
 - **BID fed**: Two times daily with meals

Safety, PK, Pharmacodynamic and Efficacy Assessments

- Laboratory assessments performed weekly for first 1-3 cycles
- Samples for pharmacokinetic and pharmacodynamic analyses were collected on C1D1, C1D15 and C2D1 (C1D1 and C1D15 fasted on TID regimen)
 - A subset of TID patients received the C2D1 dose with food to compare fed vs fasting PK at steady state
- Computed tomography (CT) scans performed at baseline and on D1 of every third cycle (starting on C4D1)

STUDY STATUS

- As of October 1, 2015, 98 patients had been treated with single agent CB-839 and had data in the clinical database
- 600 mg po BID fed was selected for expansion cohorts based on:
 - PK consistently above target threshold
 - Clear PK/pharmacodynamic relationship in platelets
 - Evidence of pharmacodynamic activity in tumors
- An MTD has not been established
 - 800mg po BID fed also considered safe and tolerable
- Summaries of patient enrollment and baseline characteristics are provided in Tables 1 and 2, respectively

Table 1: Enrollment Summary

TID Dose Escalation:		BID fed Dose Escalation and Expansion:	
100 – 800 mg		600 – 1000 mg^	
Total subjects: 32*		Total subjects: 66	
Dose level	#	Tumor type	#
100 mg	3	RCC	16
150 mg	4	TNBC	16
250 mg	11	TCA cycle mutations	23
400 mg	4	– *IDH-mutant (non-glioma)* – *SDH-deficient* – *FH-deficient*	*14* *5* *4*
600 mg	7		
800 mg	3	NSCLC	8
*Tumor types: TNBC (11), RCC (4), NSCLC (4), Mesothelioma (3), TCA cycle mutations (2), Other (8)		Mesothelioma	3
		^Dose levels: 600 mg (50), 800 mg (11), 1000 mg (5)	

** IDH 1 or 2 mutant, FH-deficient, or SDH-deficient solid tumors*

Table 2: Baseline Characteristics

		All subjects (N=98)	Select BID Expansion cohorts: RCC (N=16)	TNBC (N=16)	NSCLC (N=8)
Age: median (range)		60 (19–93	62.5 (46–93)	54.5 (36–70)	68.5 (51–76)
Female/Male: N (%)		62 (63%)/ 36 (37%)	5 (31%)/ 11 (69%)	16 (100%)/ 0 (0%)	6 (75%)/ 2 (25%)
Prior therapies (locally advanced or metastatic disease)	Median (range)	3 (0–14)	4.5 (1–9)	4.5 (1–9)	4 (1–9)
	0-1: N (%)	22 (22%)	3 (19%)	3 (19%)	2 (25%)
	2-4: N (%)	44 (45%)	5 (31%)	5 (31%)	3 (28%)
	≥5:N (%)	32 (33%)	8 (50%)	8 (50%)	3 (28%)
ECOG Score: N (%)	0	25 (27%)	4 (25%)	7 (44%)	1 (13%)
	1	69 (70%)	11 (69%)	9 (56%)	7 (88%)

PHARMACOKINETICS

- CB-839 has good PK properties in cancer patients
 - Half-life approximately 4 hr
 - Exposure generally increases with dose
 - Moderate accumulation ratio observed at steady state (median of 2.4 on TID schedule)
- Dosing with food enabled switch to BID dosing regimen
 - Positive food effect increases exposure with fed regimen (Figures 3 and 4)
 - Delayed absorption resulted in a "flatter" PK profile
 - Similar C_{max} and C_{min} with BID fed vs. TID regimen (Table 3) despite:
 - Longer dosing interval (12 hr vs. 8 hr)
 - 50% less daily dose

Figure 3: Increase in Exposure with Food

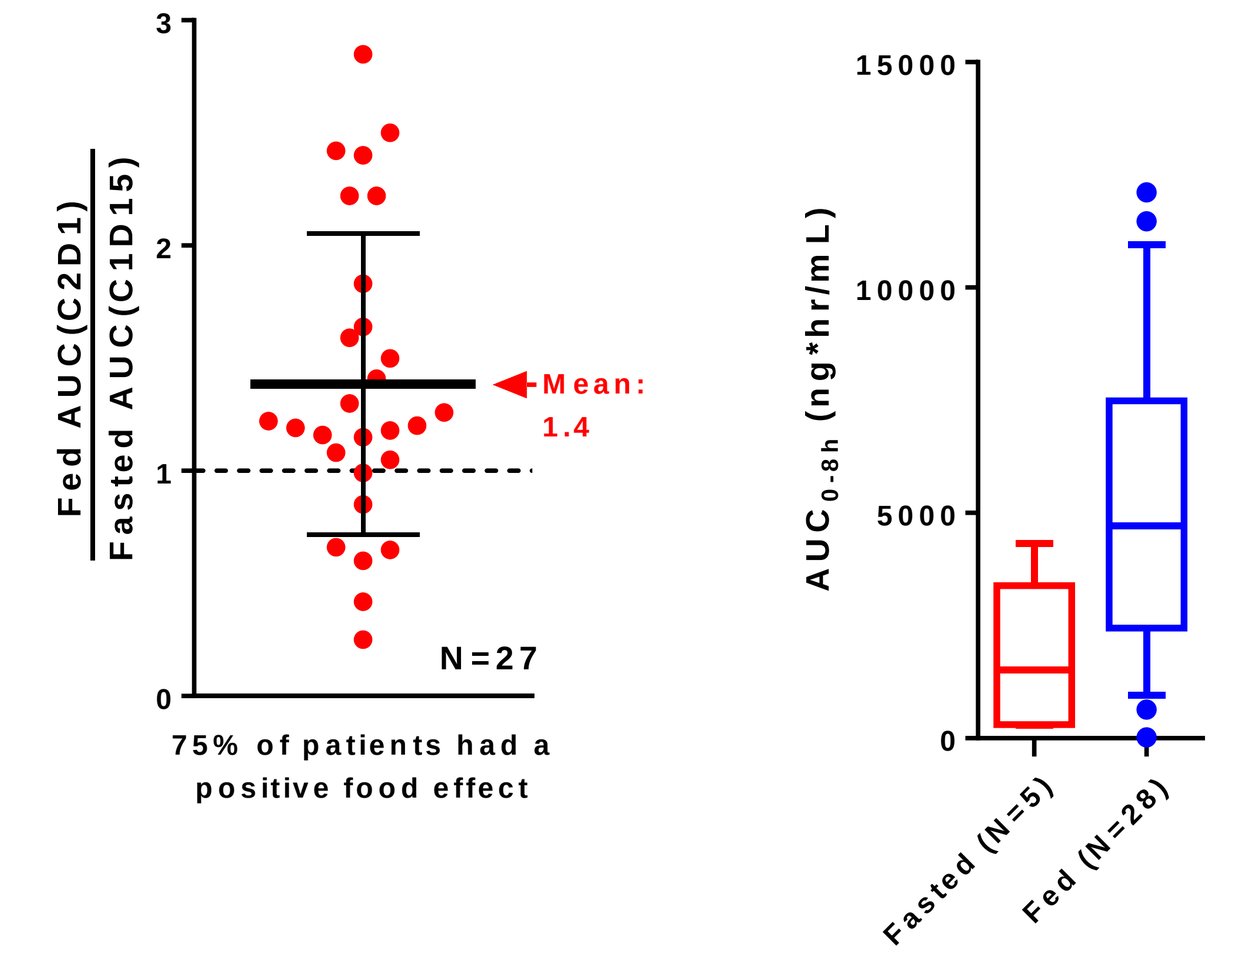
Steady State Fed vs Fasting AUC on TID Schedule
Fed AUC(C2D1) / Fasted AUC(C1D15)
Mean: 1.4
N=27
75% of patients had a positive food effect
First Dose Fed vs Fasting at 600 mg
AUC0-8h (ng*hr/mL)
Fasted (N=5)
Fed (N=28)

Table 3: PK Parameters on Fed vs Fasted Schedules

	Daily dose (mg)	Dosing interval (hr)	C1D15 PK Parameter: Mean (±SD) C_{min} (ng/mL)	C_{max} (ng/mL)	$AUC_{0\text{-}8h}$ (ng*hr/mL)^
600 mg BID fed (N=23)	1200	~12	457 (±335)	1476 (±676)	7416 (±3757)
600 mg TID (N=5)	1800	~8	495 (±390)	1565 (±922)	7344 (±4058)

^Note: $AUC_{0\text{-}8h}$ does not fully cover dosing interval on BID schedule

SAFETY AND TOLERABILITY

- Most Adverse Events (AE) have been Gr1/2 and reversible (Table 4)
 - Most frequent events were fatigue, GI events and mild LFT elevations
- Among patients receiving BID fed regimen 4.5% (3/66) experienced a Gr3/4 AE suspected to be related to CB-839 (Table 5)
- 3.0% (2/66) discontinued due to a drug-related AE (one each at 800 mg and 1000 mg)
- BID fed regimen substantially reduced rate of Gr3 LFT elevations (Table 5)
 - 1.5% (BID fed) vs 16% (TID)

Table 4: Adverse Events Occurring in >10% of Patients on the BID schedule

All AEs in ≥7 subjects on BID schedule	BID (N=66)		TID (N=32)	
MedDRA Preferred Term	Total N (%)	Drug Related N (%)	Total N (%)	Drug Related N (%)
Patients with any AE	63 (96)	44 (67)	30 (94)	23 (72)
Fatigue	19 (29)	16 (24)	15 (47)	8 (25)
Nausea	17 (26)	12 (18)	7 (22)	2 (6.3)
Constipation	10 (15)	1 (1.5)	2 (6.3)	2 (6.3)
Alanine aminotransferase increased	9 (14)	8 (12)	7 (22)	6 (19)
Anemia	9 (14)	1 (1.5)	2 (6.3)	0
Insomnia	9 (14)	1 (1.5)	1 (3.1)	0
Vomiting	9 (14)	3 (4.5)	9 (28)	4 (13)
Blood creatinine increased	8 (12)	3 (4.5)	2 (6.3)	1 (3.1)
Photophobia	8 (12)	7 (11)	1 (3.1)	1 (3.1)
Aspartate aminotransferase increased	7 (11)	7 (11)	7 (22)	6 (19)
Blood alkaline phosphatase increased	7 (11)	3 (4.5)	4 (13)	2 (6.3)
Decreased appetite	7 (11)	5 (7.6)	3 (9.4)	0
Gamma-glutamyltransferase increased	7 (11)	6 (9.1)	2 (6.3)	1 (3.1)

Table 5: Grade 3 or Higher Adverse Events Occurring in ≥2 Patients

≥Gr3 AEs in ≥2 subjects on BID schedule	BID (N=66)		TID (N=32)	
MedDRA Preferred Term	Total N (%)	Drug Related N (%)	Total N (%)	Drug Related N (%)
Patients with any ≥Gr3 AE	22 (33)	3 (4.5)	11 (34)	7 (22)
Fall	3 (4.5)	0	0	0
Anemia	2 (3.0)	0	1 (3.1)	0
Gamma-glutamyltransferase increased	2 (3.0)	2 (3.0)	2 (6.3)	1 (3.1)
Pneumonia	2 (3.0)	0	1 (3.1)	0
Syncope	2 (3.0)	0	0	0
All ≥Gr3 LFT AEs				
Alanine aminotransferase increased	1 (1.5)	1 (1.5)	5 (16)	5 (16)
Aspartate aminotransferase increased	0	0	6 (19)	5 (16)
Blood alkaline phosphatase increased	1 (1.5)	0	3 (9.4)	1 (3.1)
Blood bilirubin increased	1 (1.5)	0	1 (3.1)	0

CLINICAL OUTCOMES

- This Phase 1 study has enrolled patients with a variety of tumor types across a range of doses on two different dosing regimens [BID fed (n=66) and TID (n=32)]
- 44% of patients on BID fed regimen have achieved a PR or SD as best response (Table 6)
- 60% of RCC patients on the BID schedule achieved a PR or SD (Table 7)
- Five patients remained on CB-839 for >8 months without progression (Figures 4)
 - 2 TNBC, 1 RCC, 1 mesothelioma, 1 IDH1 mutant chondrosarcoma

Table 6: Best Overall Response

	BID (N=66)	TID (N=32)
RECIST Response Evaluable (N)	50	27
PR	1 (2%)	
SD	21 (42%)	6 (22%)
PD	26	19
Disease-related death prior to scan	1	2
Withdrew for drug-related AE prior to scan	1	
Not evaluable (N)	16	5
On study, prior to first tumor assessment	1	
Discontinued before tumor assessment	15	5

Table 7: Response by Tumor Type

Disease Type	BID Schedule: Response Evaluable	SD	PR	% SD or better	TID Schedule: Response Evaluable	SD	PR	% SD or better
RCC	15	8	1	60	4	2		50
TNBC	9	1		11	10	2		20
TCA cycle mutant	18	8		44	1			
– *IDH-mutant*	*11*	*4*		*36*				
– *SDH deficient*	*3*	*2*		*67*	*1*			
– *FH-deficient*	*4*	*2*		*50*				
NSCLC	6	2		33	3	1		33
Mesothelioma	2	2		100	2	1		50
Other					7			
Total	50	21	1	44	27	6		22

Figure 4: Treatment Duration

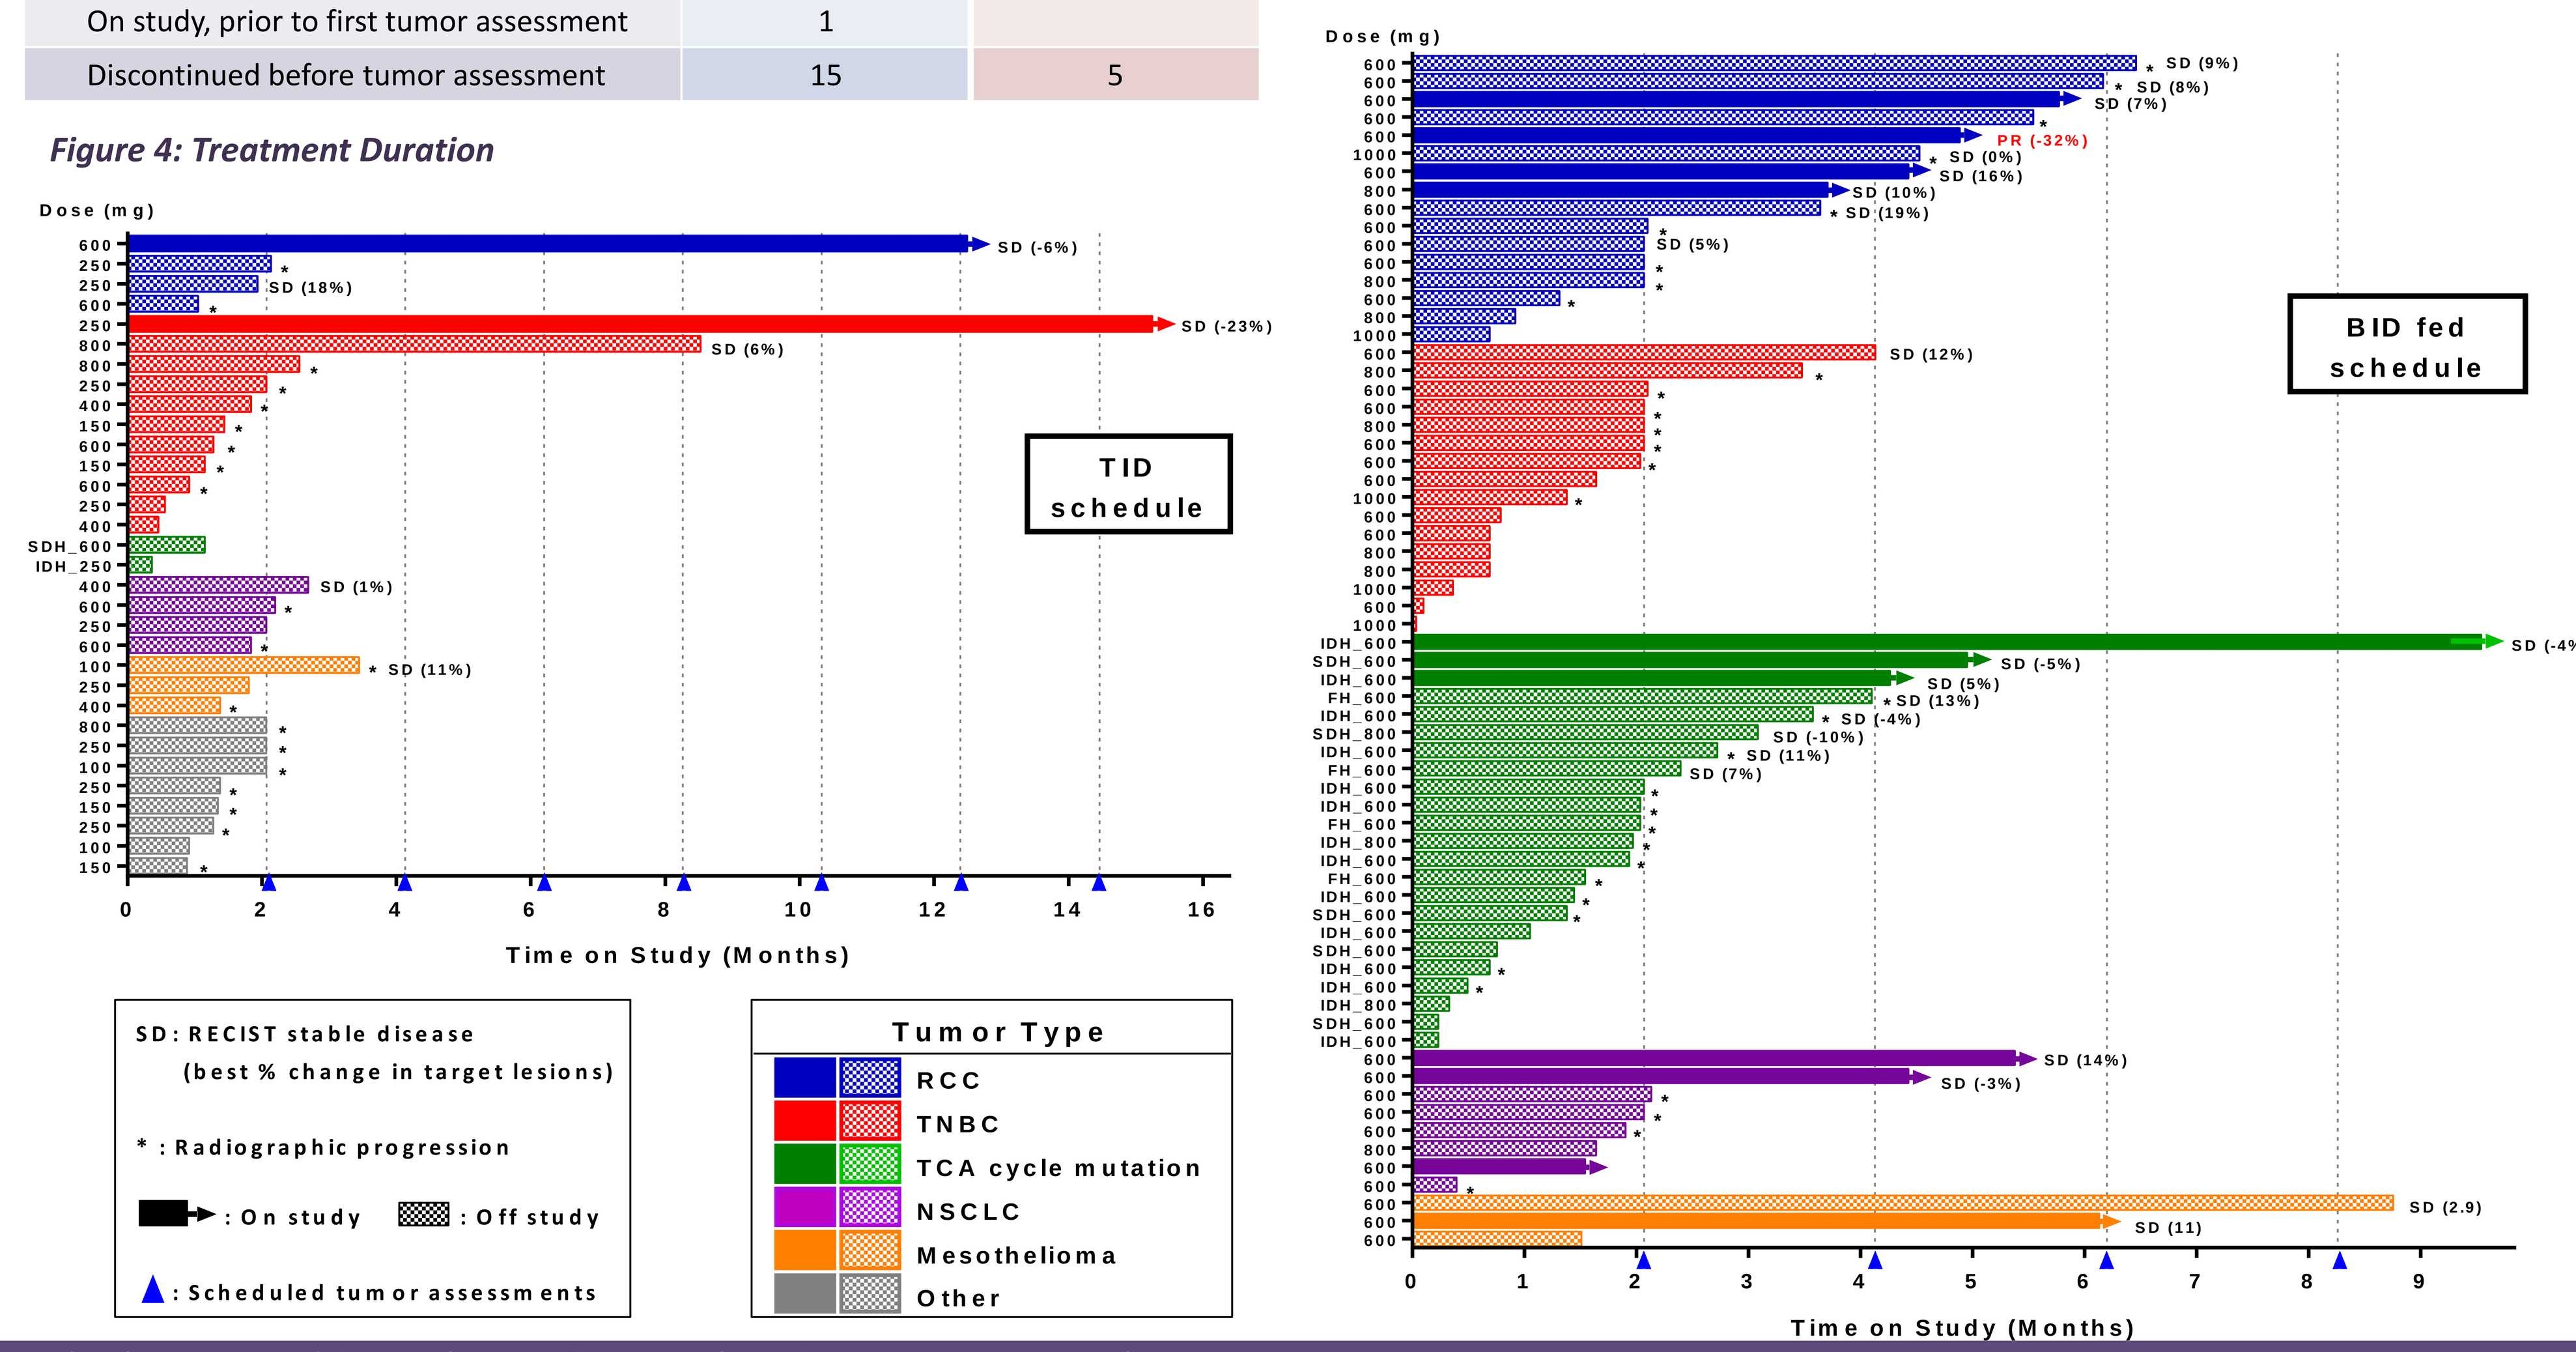
Dose (mg)
TID schedule
SD (-6%)
SD (18%)
SD (-23%)
SD (6%)
SD (1%)
SD (11%)
Time on Study (Months)
SD: RECIST stable disease (best % change in target lesions)
*: Radiographic progression
: On study
: Off study
: Scheduled tumor assessments
Tumor Type
RCC
TNBC
TCA cycle mutation
NSCLC
Mesothelioma
Other
BID fed schedule
SD (9%)
SD (8%)
SD (7%)
PR (-32%)
SD (0%)
SD (16%)
SD (10%)
SD (19%)
SD (5%)
SD (12%)
SD (-4%)
SD (-5%)
SD (5%)
SD (13%)
SD (-4%)
SD (-10%)
SD (11%)
SD (7%)
SD (14%)
SD (-3%)
SD (2.9)
SD (11)
Time on Study (Months)

EFFICACY IN RENAL CELL CARCINOMA

- Sixteen RCC patients enrolled on BID fed regimen
 - 9 clear cell, 4 papillary, 3 with other histologies
- Nine of 15 evaluable patients had stable disease (SD) or better as best response
 - Eight stable disease and one PR for a Clinical Benefit Rate (CBR) of 60%
- Median time on study for 16 RCC patients on BID fed schedule ~4 months
 - Compares favorably with historical everolimus data in earlier stage Ph3 RCC studies (Table 7)

Table 7: Response Rates and PFS from Everolumus Studies in RCC

Treatment	Study	# Prior lines (median)	ORR	CBR	PFS
Everolimus	Motzer, 2015[5]	≥1 (1)	5%	NR	4.4m
Everolimus	Choueiri, 2015[6]	1-2 (1)	5%	67%	3.8m
Everolimus	Motzer, 2010[7]	1-2 (1)	1.8%	68.6%	4.9m
Placebo	Motzer, 2010[7]	1-2 (1)	0%	32.4%	1.9m

One partial response in a patient with clear cell RCC (Figure 5)

- 32% reduction in target lesions
 - Complete resolution of lymphadenopathy
 - Adrenal target lesion with apparently necrotic center unchanged
- Disease history
 - Diagnosed with metastatic clear cell RCC in 2012
 - Three prior lines of systemic therapy (sunitinib, pazopanib, everolimus)
 - Most recent therapy was everolimus
 - Remained on everolimus for 4.5 months
 - Best response was progressive disease
- Relatively limited evaluation of genetic/molecular markers performed to date

Figure 5: Radiological Response to CB-839 in Clear Cell RCC Patient

Screening

Cycle 7/ Day1

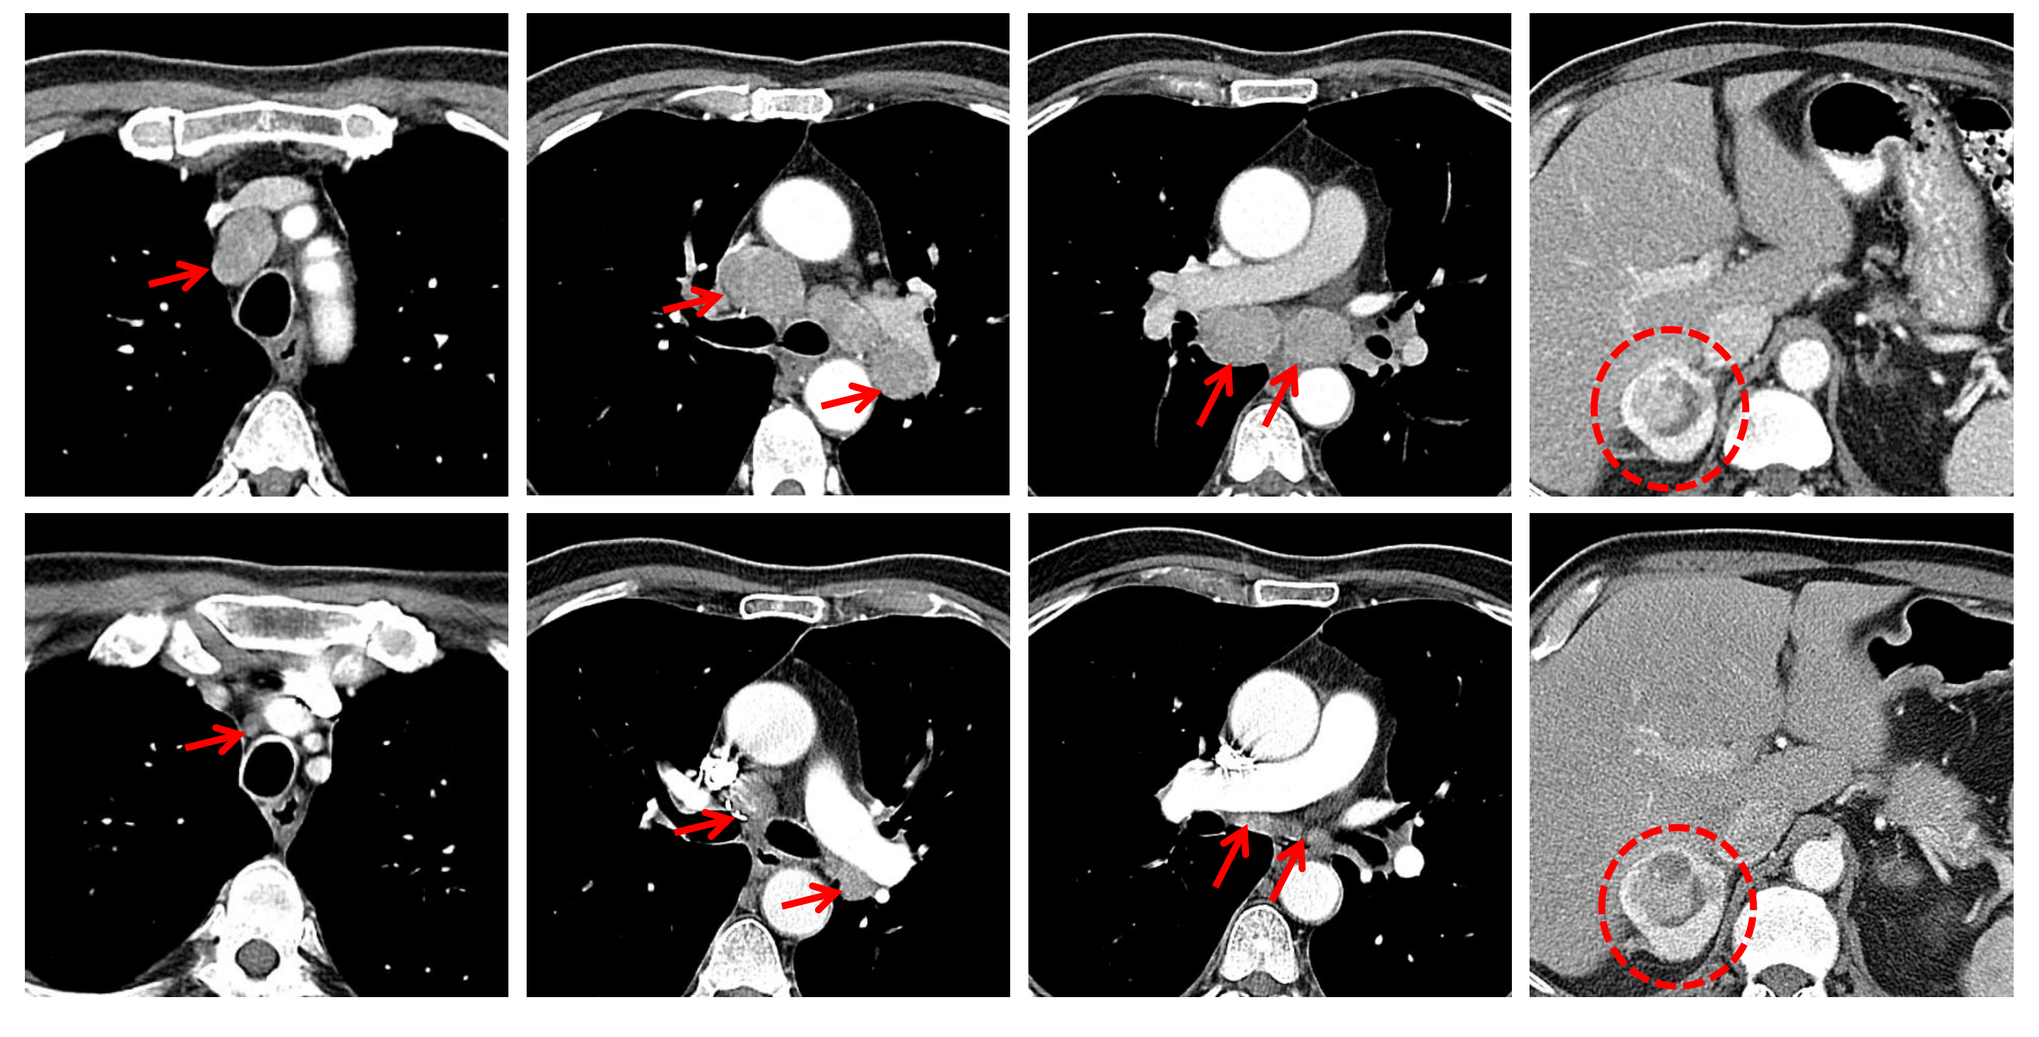

IV contrast-enhanced CT scans were performed at screening and Cycle 7/Day 1 in this patient with clear cell RCC. Near complete regression of mediastinal and hilar lymphadenopathy had occurred after treatment, whereas a complex adrenal metastatic lesion was grossly unchanged. RECIST 1.1 showed a partial response (PR) with a 32% reduction in the target lesions.

SUMMARY AND CONCLUSIONS

- **CB-839 is well tolerated in advanced cancer patients**
- **Dosing with meals allows convenient BID dosing and improved safety profile**
- **Primary efficacy signal to date has been in renal cell carcinoma**
- **Expanded development in RCC planned**
 - RCC enrollment expanded on monotherapy
 - Everolimus combination ongoing in RCC
- **Development continues in other indications**
 - Non-small cell lung cancer: erlotinib combo
 - Triple negative breast cancer: paclitaxel combo
 - AML: azacitidine combination
 - Myeloma: pomalidomide/dexamethasone combination

References

1. Wise and Thompson (2010) *Trends Biochem Sci* **35**:427-433
2. DeBerardinis and Cheng (2010) *Oncogene* **29**:313-324
3. Gross et al. (2014) *Mol Cancer Ther* **13**:890-901
4. Parlati et al. (2013) *Blood* **122**:4226
5. Motzer et al. (2015) *N Engl J Med* (in press)
6. Choueiri et al. (201) *N Engl J Med* (in press)
7. Motzer et al. (2010) Cancer **116**:4256

2015 AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics, November 5-9, 2015, Boston, Massachusetts