CB-839, a novel potent and selective glutaminase inhibitor, has broad antiproliferative activity in cell lines derived from both solid tumors and hematological malignancies

Francesco Parlati, Susan Demo, Matthew Gross, Julie Janes, Evan Lewis, Andy MacKinnon, Mirna Rodriguez, Peter J. Shwonek, Taotao Wang, Jinfu Yang, Winter Zhang, Frances Zhao and Mark Bennett

Calithera Biosciences Inc., South San Francisco, CA

Abstract

Many tumor cells utilize altered metabolic pathways to meet the bioenergetic and biosynthetic demands of rapid and sustained growth. One of the key nutrients that fuels tumor growth is the amino acid glutamine. It has long been recognized that the growth and survival of many tumor cell lines *in vitro* is dependent on extracellular glutamine. A critical step in the utilization of glutamine is its conversion to glutamate by the mitochondrial enzyme glutaminase. Glutamate and glutamate-derived metabolites in turn support a number of crucial cellular pathways including the citric acid cycle, redox balance and amino acid synthesis. CB-839 is a novel and selective inhibitor of glutaminase that has antitumor activity in preclinical models of triple negative breast cancer (TNBC), a tumor type that is particularly dependent on glutamine. Across a panel of breast cancer cell lines, the activity of CB-839 correlates with high expression of glutaminase, specifically the GAC splice variant but not the KGA splice variant, and low expression of glutamine synthetase (GLUL), an enzyme that converts glutamate to glutamine. This expression pattern is found in primary TNBC tumors suggesting a reliance on exogenous glutamine and glutaminase activity *in vivo*. To determine if other tumor types have a similar expression pattern, we undertook a systematic evaluation of GAC, KGA and GLUL expression across a diverse set of primary tumors using a normalized microarray dataset allowing comparison across a range of tumor and normal tissue samples. Elevated GAC and decreased GLUL expression relative to other tumor types or corresponding normal tissue was identified in a number of tumor types including non-small cell lung cancer, multiple myeloma, and non-Hodgkin's lymphoma. To explore whether this expression pattern predicts a reliance on glutamine and glutaminase, we tested the glutamine dependence and the activity of CB-839 on a panel of 72 cell lines representing 5 tumor types indicated by the primary tumor expression analysis. Across this cell line panel, the majority were dependent on glutamine showing either cell death, growth arrest or slowed growth after glutamine withdrawal. Similarly, CB-839 had antiproliferative activity in the majority of cell lines with IC50s in the range of 10-300 nM. Importantly, there was a strong correlation between glutamine dependence and response to 1 μM CB-839 as measured by relative cell growth or death (correlation coefficient 0.72, p<0.0001), suggesting that the glutamine requirement for tumor cell growth or survival is mediated by glutaminase and that this phenotype is consistent across numerous tumor types. These data suggest that CB-839 may have therapeutic activity in a range of tumor types that are glutamine dependent and motivate the clinical evaluation of this compound targeting a novel tumor survival pathway.

Introduction

Many tumors rely on the catabolism of glutamine to produce metabolic intermediates that fuel bioenergetic and biosynthetic demands (Ref 1, 2). Glutaminase initiates this process by converting glutamine to glutamate which is subsequently used in multiple reactions that support tumor cell growth and survival, including the generation of energy (TCA cycle), synthesis of amino acids and production of glutathione.

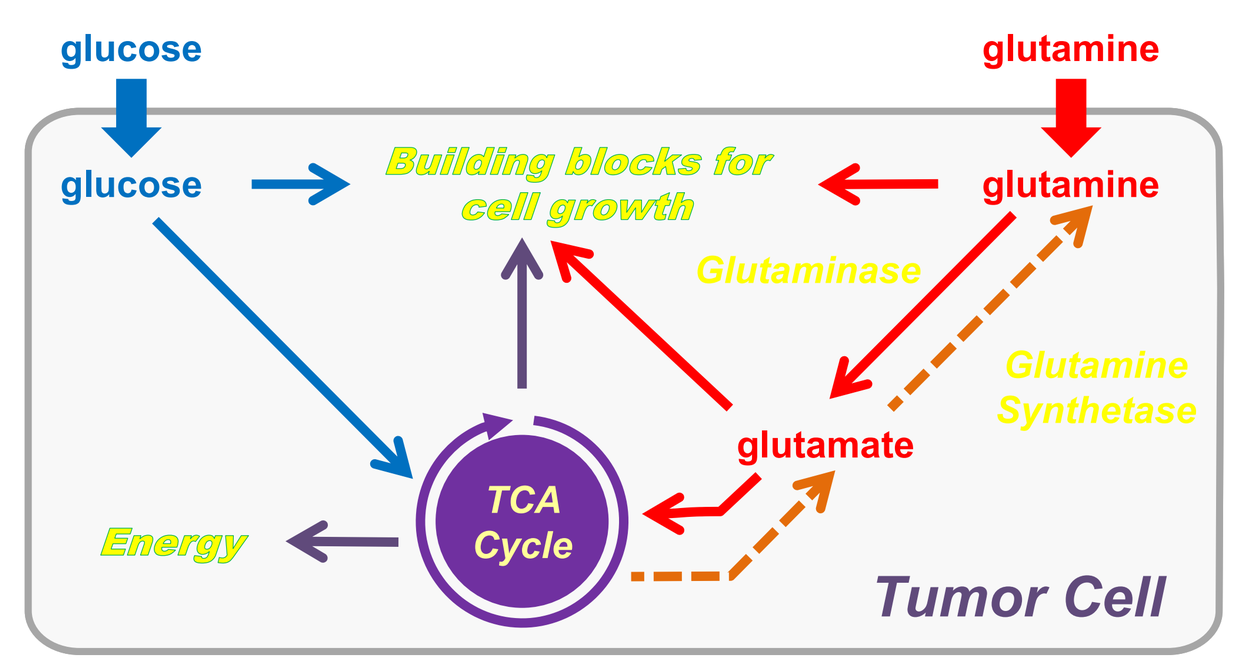

glucose
glutamine
glucose
Building blocks for cell growth
glutamine
Glutaminase
Glutamine Synthetase
glutamate
TCA Cycle
Energy
Tumor Cell

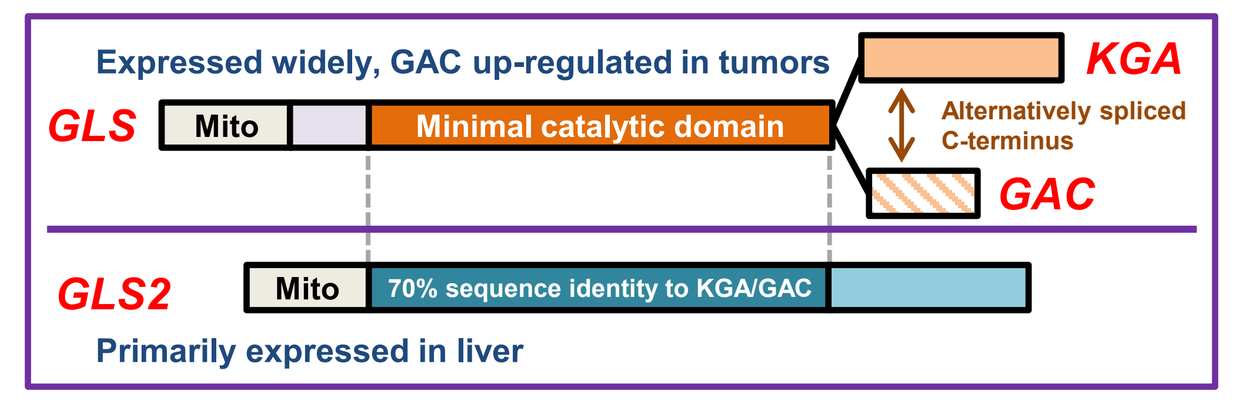

Expressed widely, GAC up-regulated in tumors
KGA
GLS
Mito
Minimal catalytic domain
Alternatively spliced C-terminus
GAC
GLS2
Mito
70% sequence identity to KGA/GAC
Primarily expressed in liver

Two human genes encode glutaminase proteins, GLS (broadly expressed) and GLS2 (primarily expressed in liver). The GLS gene produces two alternatively spliced variants, KGA and GAC, that function as homo-tetrameric enzymes. Inhibition of these two forms of glutaminase with siRNA knockdown or small molecules results in anti-proliferative effects in tumor cells both *in vitro* and *in vivo* (Ref 2, 3, 4, 5). Furthermore, the KGA and GAC splice variants show altered expression patterns in tumors (Ref 5) and their expression is associated with the aggressiveness of the cancer (Ref 6).

We have developed a series of novel, potent, and selective glutaminase inhibitors that are orally bioavailable and show *in vitro* and *in vivo* anti-tumor activity (Ref 7). Our findings provide a strong preclinical rationale for the further development of glutaminase inhibitors in oncology.

CB-839 is a Potent Glutaminase Inhibitor

CB-839 chemical structure, biochemical & cell-based activity

A

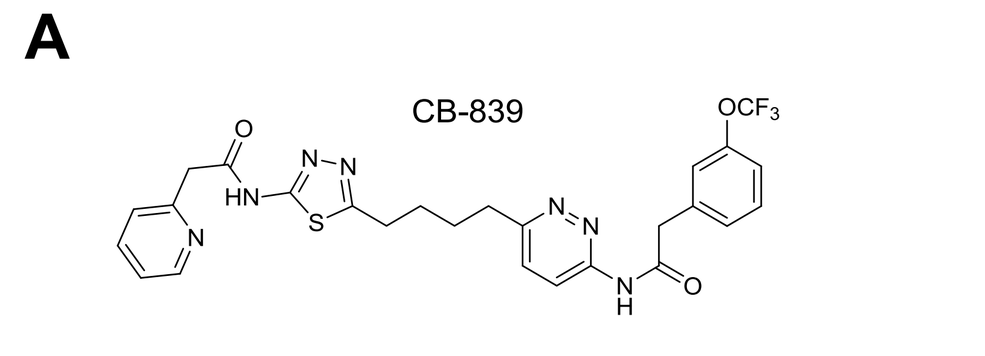

CB-839

B

Enzyme	IC_{50} (nM)
rHuGAC	29
Kidney (KGA)	24
Liver (GLS2)	>5000

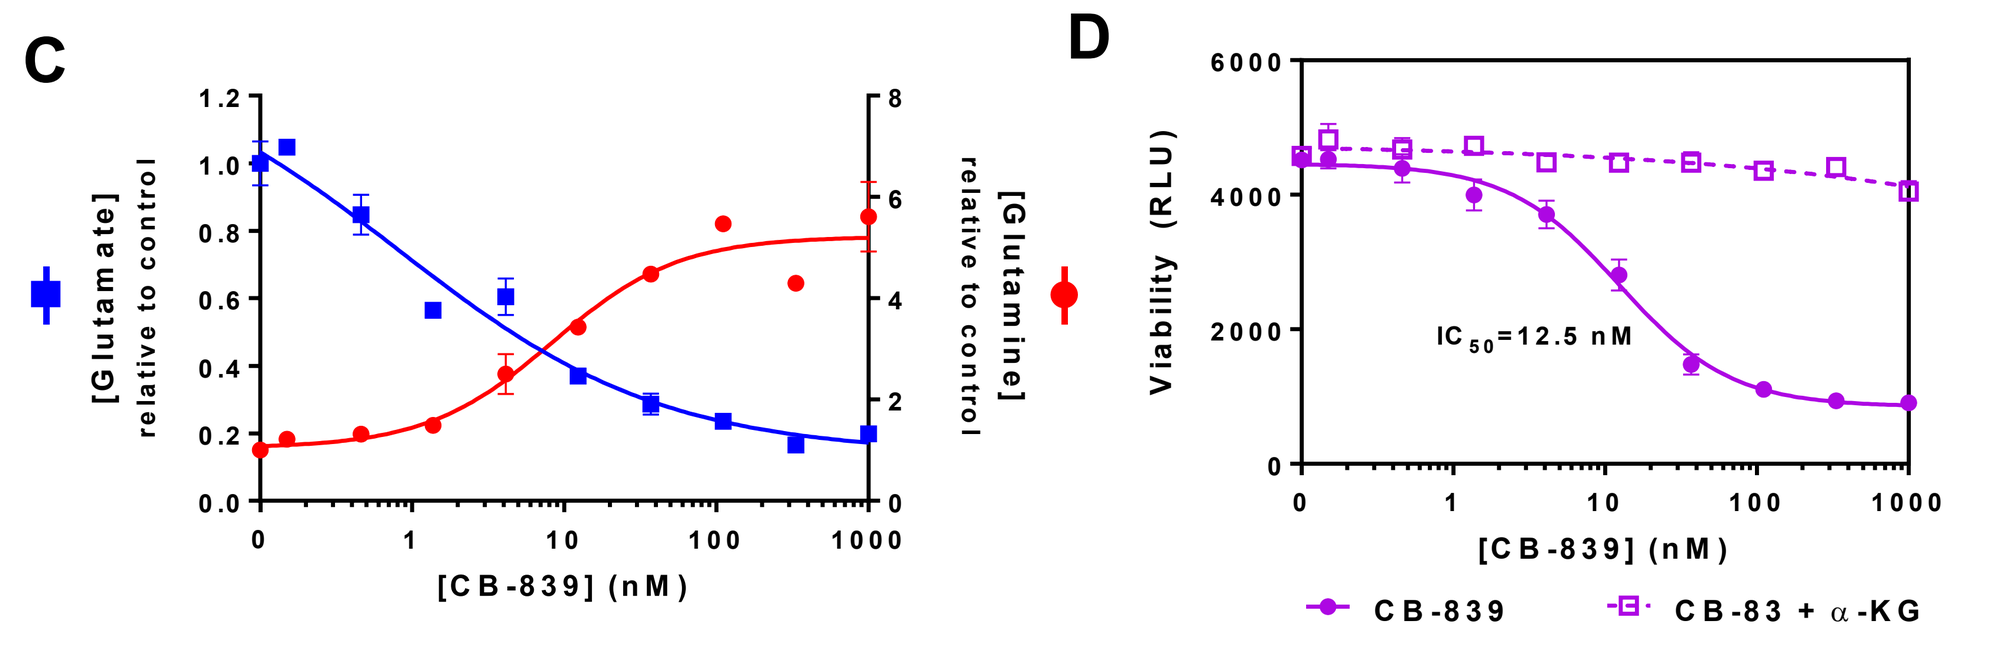

C
[Glutamate] relative to control
[Glutamine] relative to control
[CB-839] (nM)
D
Viability (RLU)
IC_{50}=12.5 nM
[CB-839] (nM)
CB-839
CB-83 + α-KG

Figure 1: **CB-839 is a potent inhibitor of glutaminase. *A*, CB-839 chemical structure. *B*, CB-839 is a potent inhibitor of both GAC and KGA splice variants of glutaminase.** Glutaminase activity was measured using a coupled glutaminase-glutamate dehydrogenase (GDH) reaction: glutaminase converts glutamine to glutamate, then GDH utilizes glutamate to convert $NADP^+$ to NADPH which is measured fluorometrically [Ex:340 nm- Em:460 nm]. CB-839 was dose titrated and incubated for 1 h with either purified recombinant human glutaminase (rHu-GAC) or lysates from mouse kidney (which expresses primarily KGA), or mouse liver (which expresses primarily GLS2). Glutamine was added to the lysates or purified enzyme after the 1 h pre-incubation and NADPH production was monitored by fluorescence for 5 min and initial velocities were calculated and normalized to uninhibited control. Percent activity was plotted against compound concentration and resulting curves were fit using a four-parameter dose response to determine IC_{50} values. ***C*, CB-839 treatment results in an increase in cellular glutamine (substrate) and a decrease in cellular glutamate (product).** MDA-MB-231 breast adenocarcinoma cells were treated with CB-839 for 24 h. Cells were harvested and the amounts of glutamine and glutamate were measured by LC/MS/MS. The measured concentrations were normalized to cell number determined on a parallel plate and plotted as a fraction relative to the DMSO control. ***D*, The anti-proliferative activity of CB-839 is correlated with changes in glutamine/glutamate levels and is reversed by addition of the downstream product α-ketoglutarate (α-KG).** MDA-MB-231 cells were treated with a range of CB-839 concentrations for 72 h in the presence or absence of 1mM di-methyl α-KG, a membrane permeable form of α-KG. The effect on cell proliferation was measured using CellTiterGlo reagent and relative luminescence units (RLU) are plotted.

Breast Cancer Biomarker Analysis

TNBC cells are responsive to CB-839 and show increased GAC expression, GLS activity and Glu:Gln ratio as compared to receptor positive breast cancer cell lines

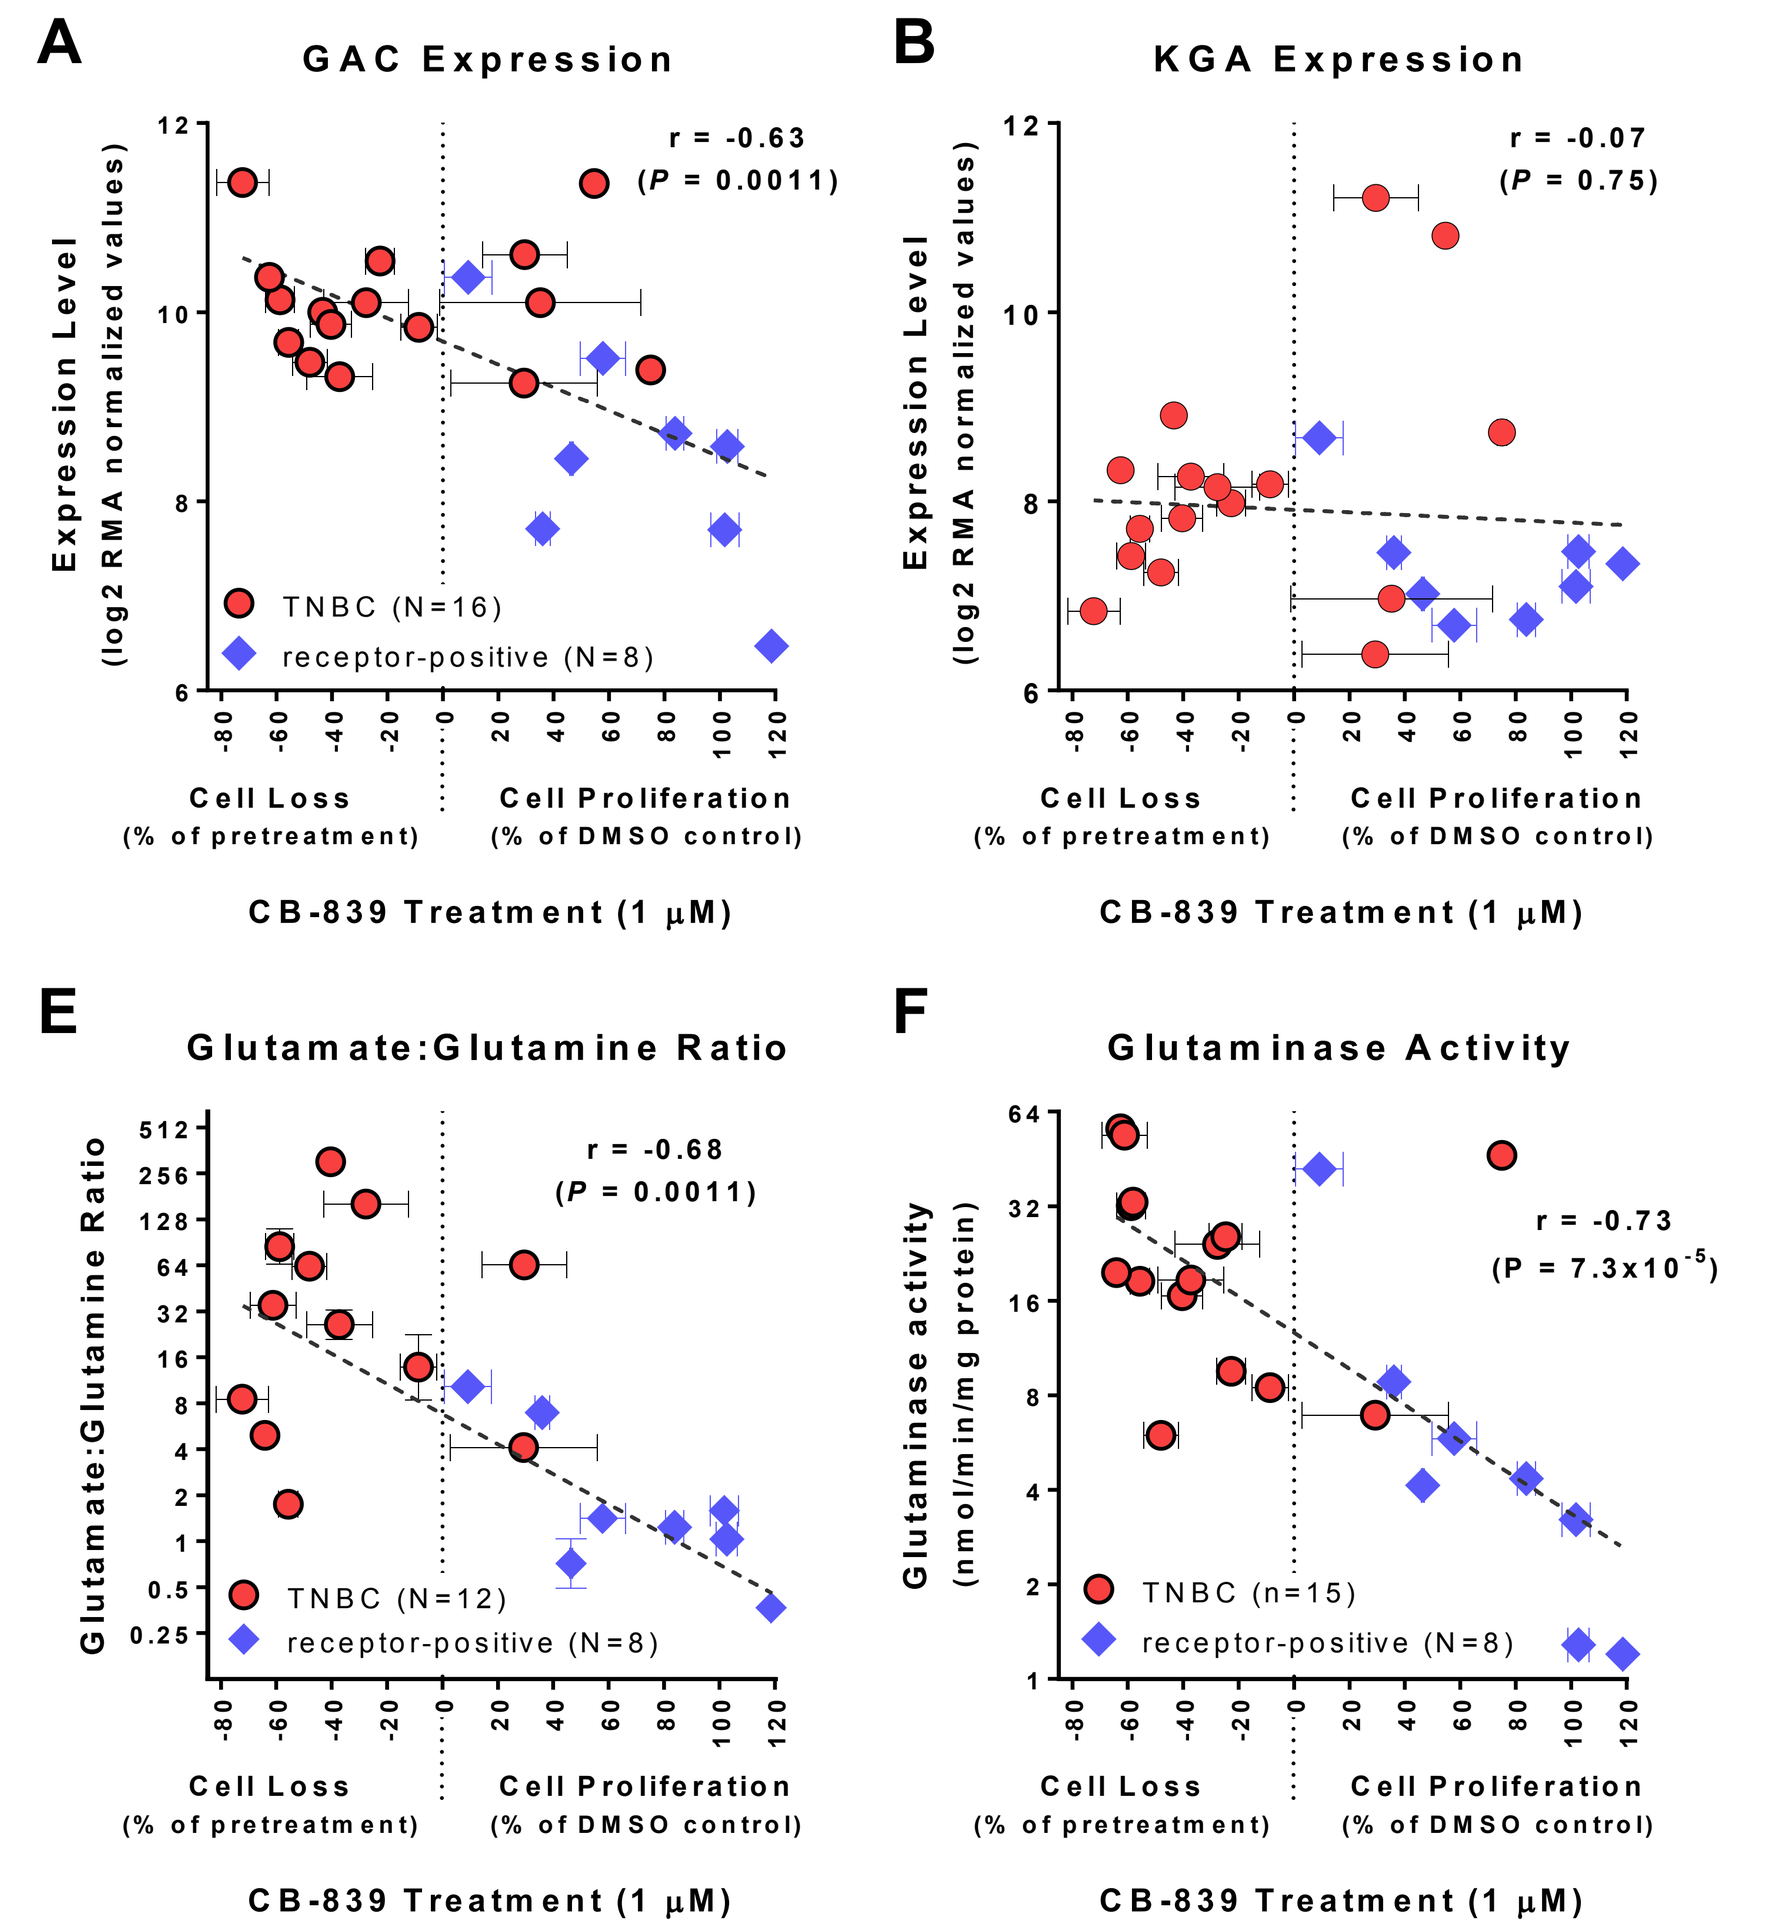

A GAC Expression
r = -0.63 (P = 0.0011)
B KGA Expression
r = -0.07 (P = 0.75)
C GLUL Expression
r = 0.39 (P = 0.06)
D GLS2 Expression
r = 0.41 (P = 0.048)
Expression Level (log2 RMA normalized values)
TNBC (N=16)
receptor-positive (N=8)
Cell Loss (% of pretreatment)
Cell Proliferation (% of DMSO control)
CB-839 Treatment (1 μM)
E Glutamate:Glutamine Ratio
r = -0.68 (P = 0.0011)
TNBC (N=12)
receptor-positive (N=8)
F Glutaminase Activity
Glutaminase activity (nmol/min/mg protein)
r = -0.73 (P = 7.3x10^-5)
TNBC (n=15)
receptor-positive (N=8)

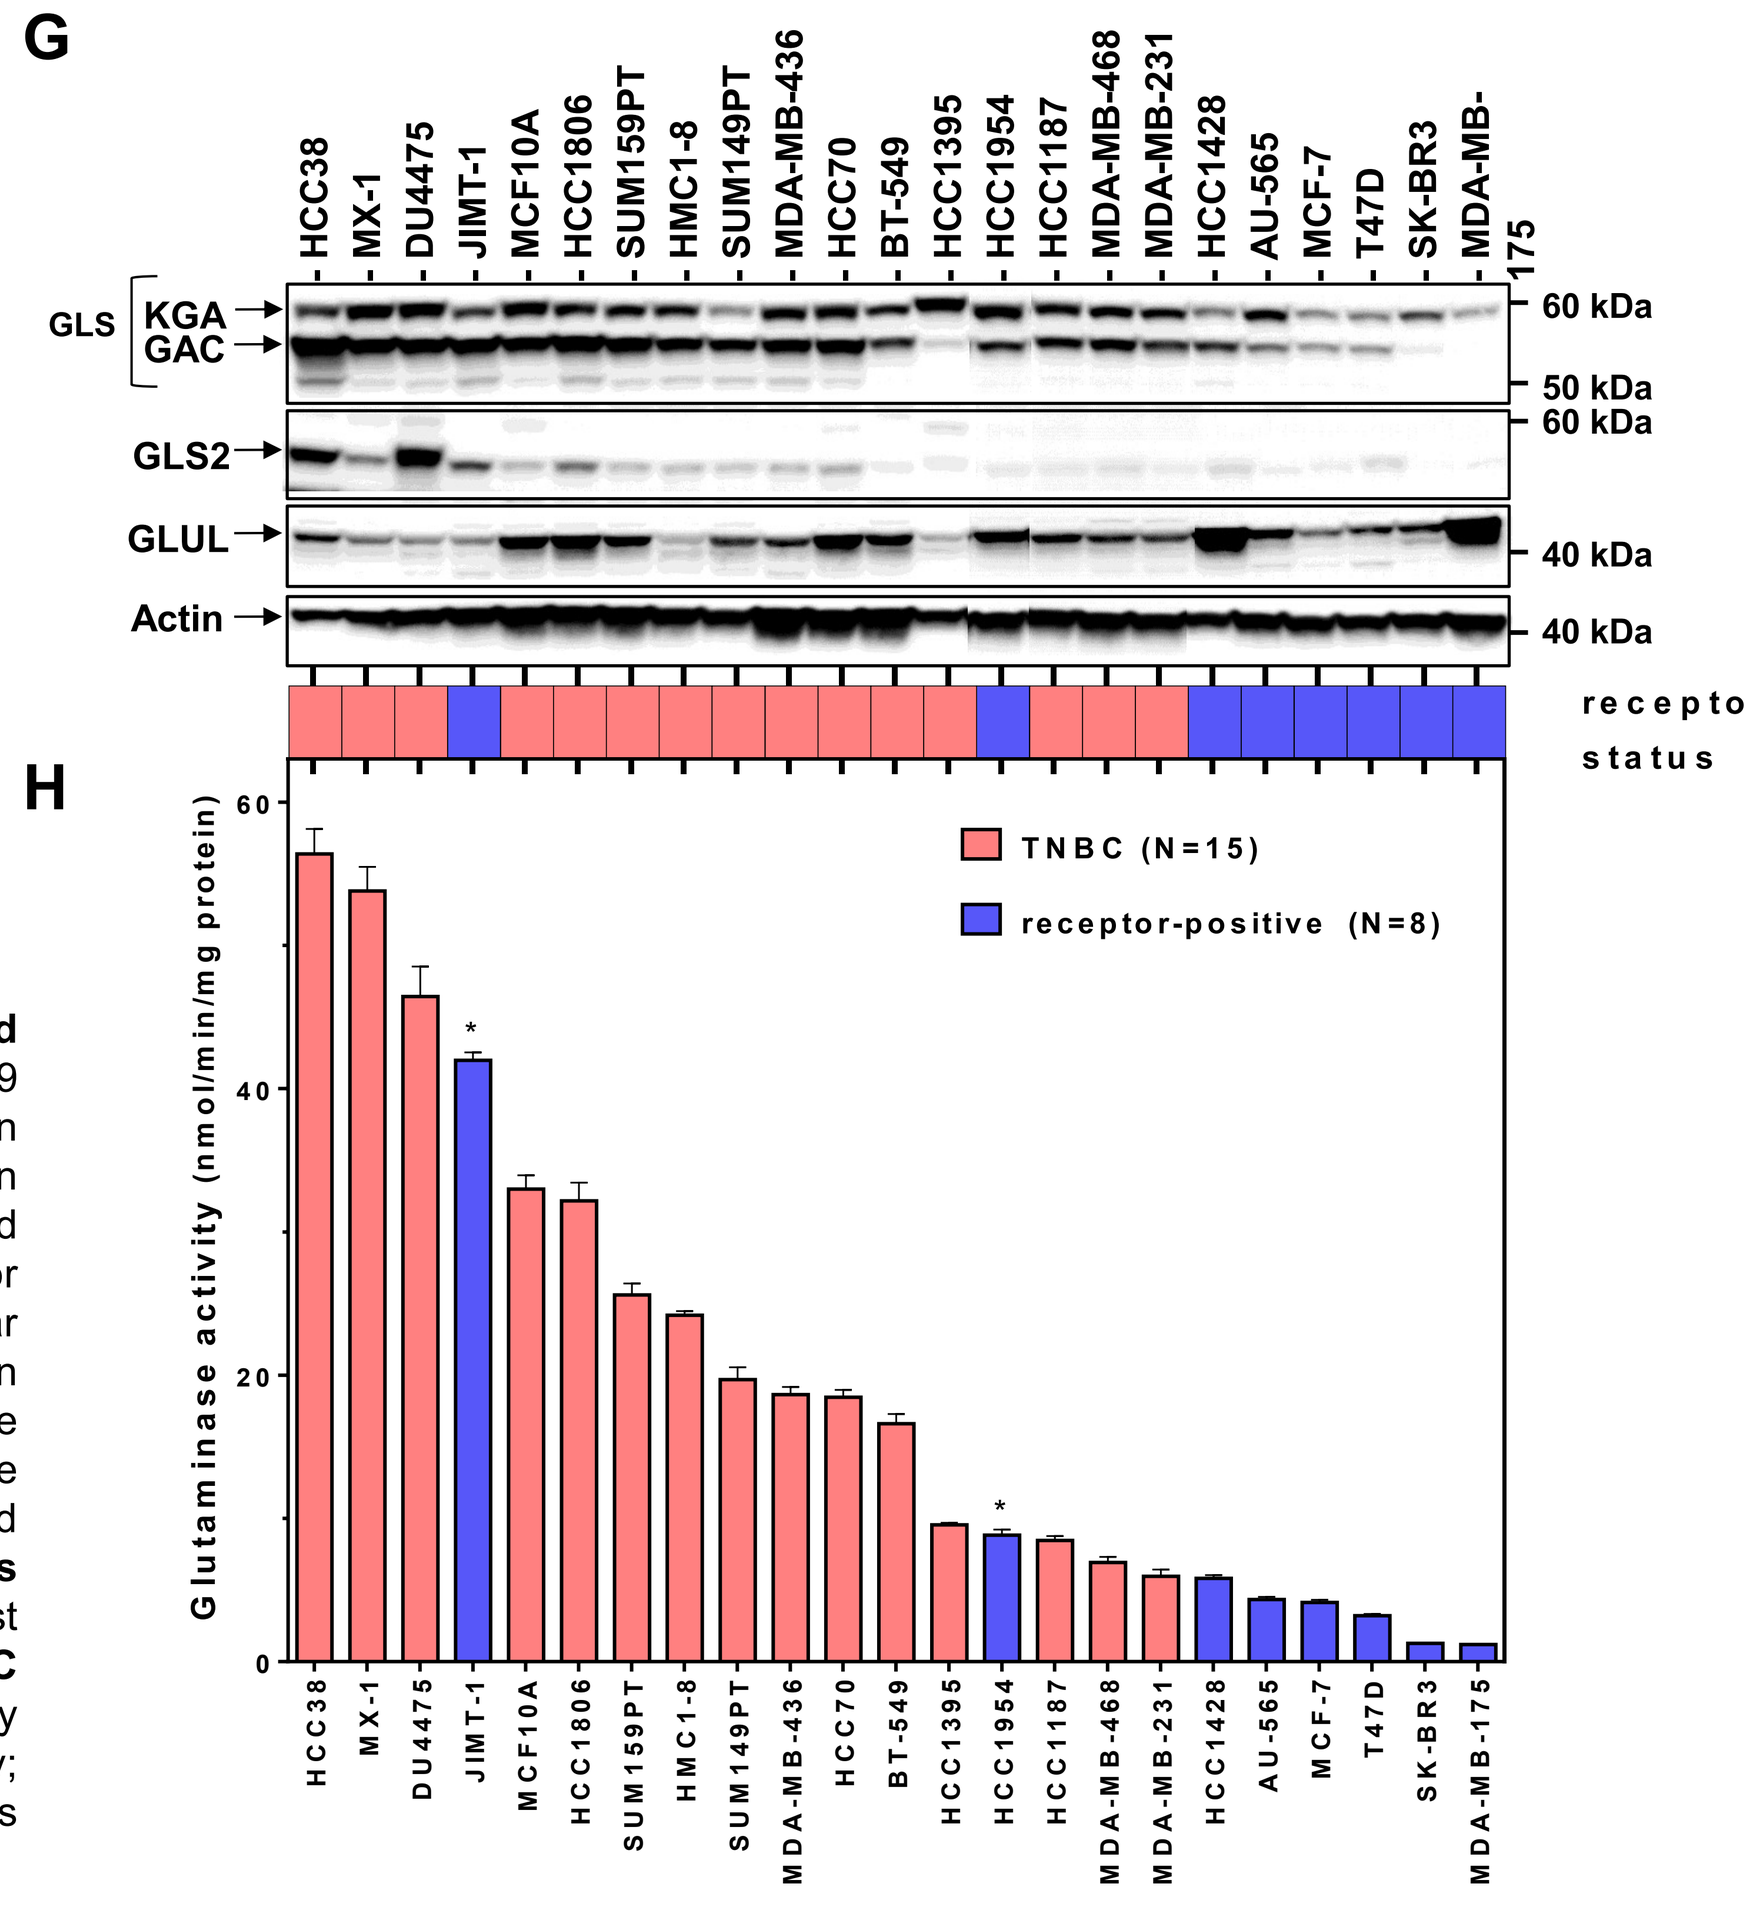

G
HCC38
MX-1
DU4475
JIMT-1
MCF10A
HCC1806
SUM159PT
HMC1-8
SUM149PT
MDA-MB-436
HCC70
BT-549
HCC1395
HCC1954
HCC1187
MDA-MB-468
MDA-MB-231
HCC1428
AU-565
MCF-7
T47D
SK-BR3
MDA-MB-175
GLS
KGA
GAC
GLS2
GLUL
Actin
60 kDa
50 kDa
60 kDa
40 kDa
40 kDa
receptor status
H
Glutaminase activity (nmol/min/mg protein)
TNBC (N=15)
receptor-positive (N=8)

Figure 2: Correlation of CB-839 sensitivity with *GAC* expression, glutaminase activity and metabolite levels. ***A-D,*** Correlation between cell proliferation or loss measured after CB-839 treatment plotted on the x-axis and *GAC (A)*, *KGA (B)*, *GLUL (C), or GLS2 (D)* mRNA expression from the CCLE database (log2 RMA normalized values) plotted on the y-axis. ***E***, Correlation between cell proliferation or loss measured after CB-839 treatment on the x-axis and glutaminase specific activity plotted on the y-axis. ***F*,** Correlation between cell proliferation or loss measured after CB-839 treatment plotted on the x-axis and the baseline ratio of intracellular glutamate to glutamine plotted on the y-axis. For all panels, each data point depicts the mean and SEM from at least duplicate measurements for an individual cell line. The zero value on the x-axis represents the cell density at the time of compound addition and the dashed line is the linear regression fit [using log2 transformed values in panels (C) and (*D*)]. The calculated Pearson correlation coefficients (r) and associated *P* values are shown. ***G*, TNBC cell lines have elevated glutaminase protein levels.** SDS-PAGE and immunoblot analyses of a breast cancer cell line panel using antibodies that recognize GAC, KGA, GLS2, and GLUL. ***H*, TNBC cell lines have elevated glutaminase activity levels.** Phosphate-activated glutaminase activity was measured in cell line homogenates and normalized to protein amount used in the assay; mean and SEM from duplicate measurements are plotted. The basal-like ER-/HER2+ cell lines JIMT-1 and HCC1954 are annotated with asterisks (*).

Potential Clinical Biomarkers in TNBC

Primary TNBC tumors show increased expression of GAC mRNA, increased glutamate:glutamine and increased glutaminase protein expression as compared to primary ER^+ breast tumors.

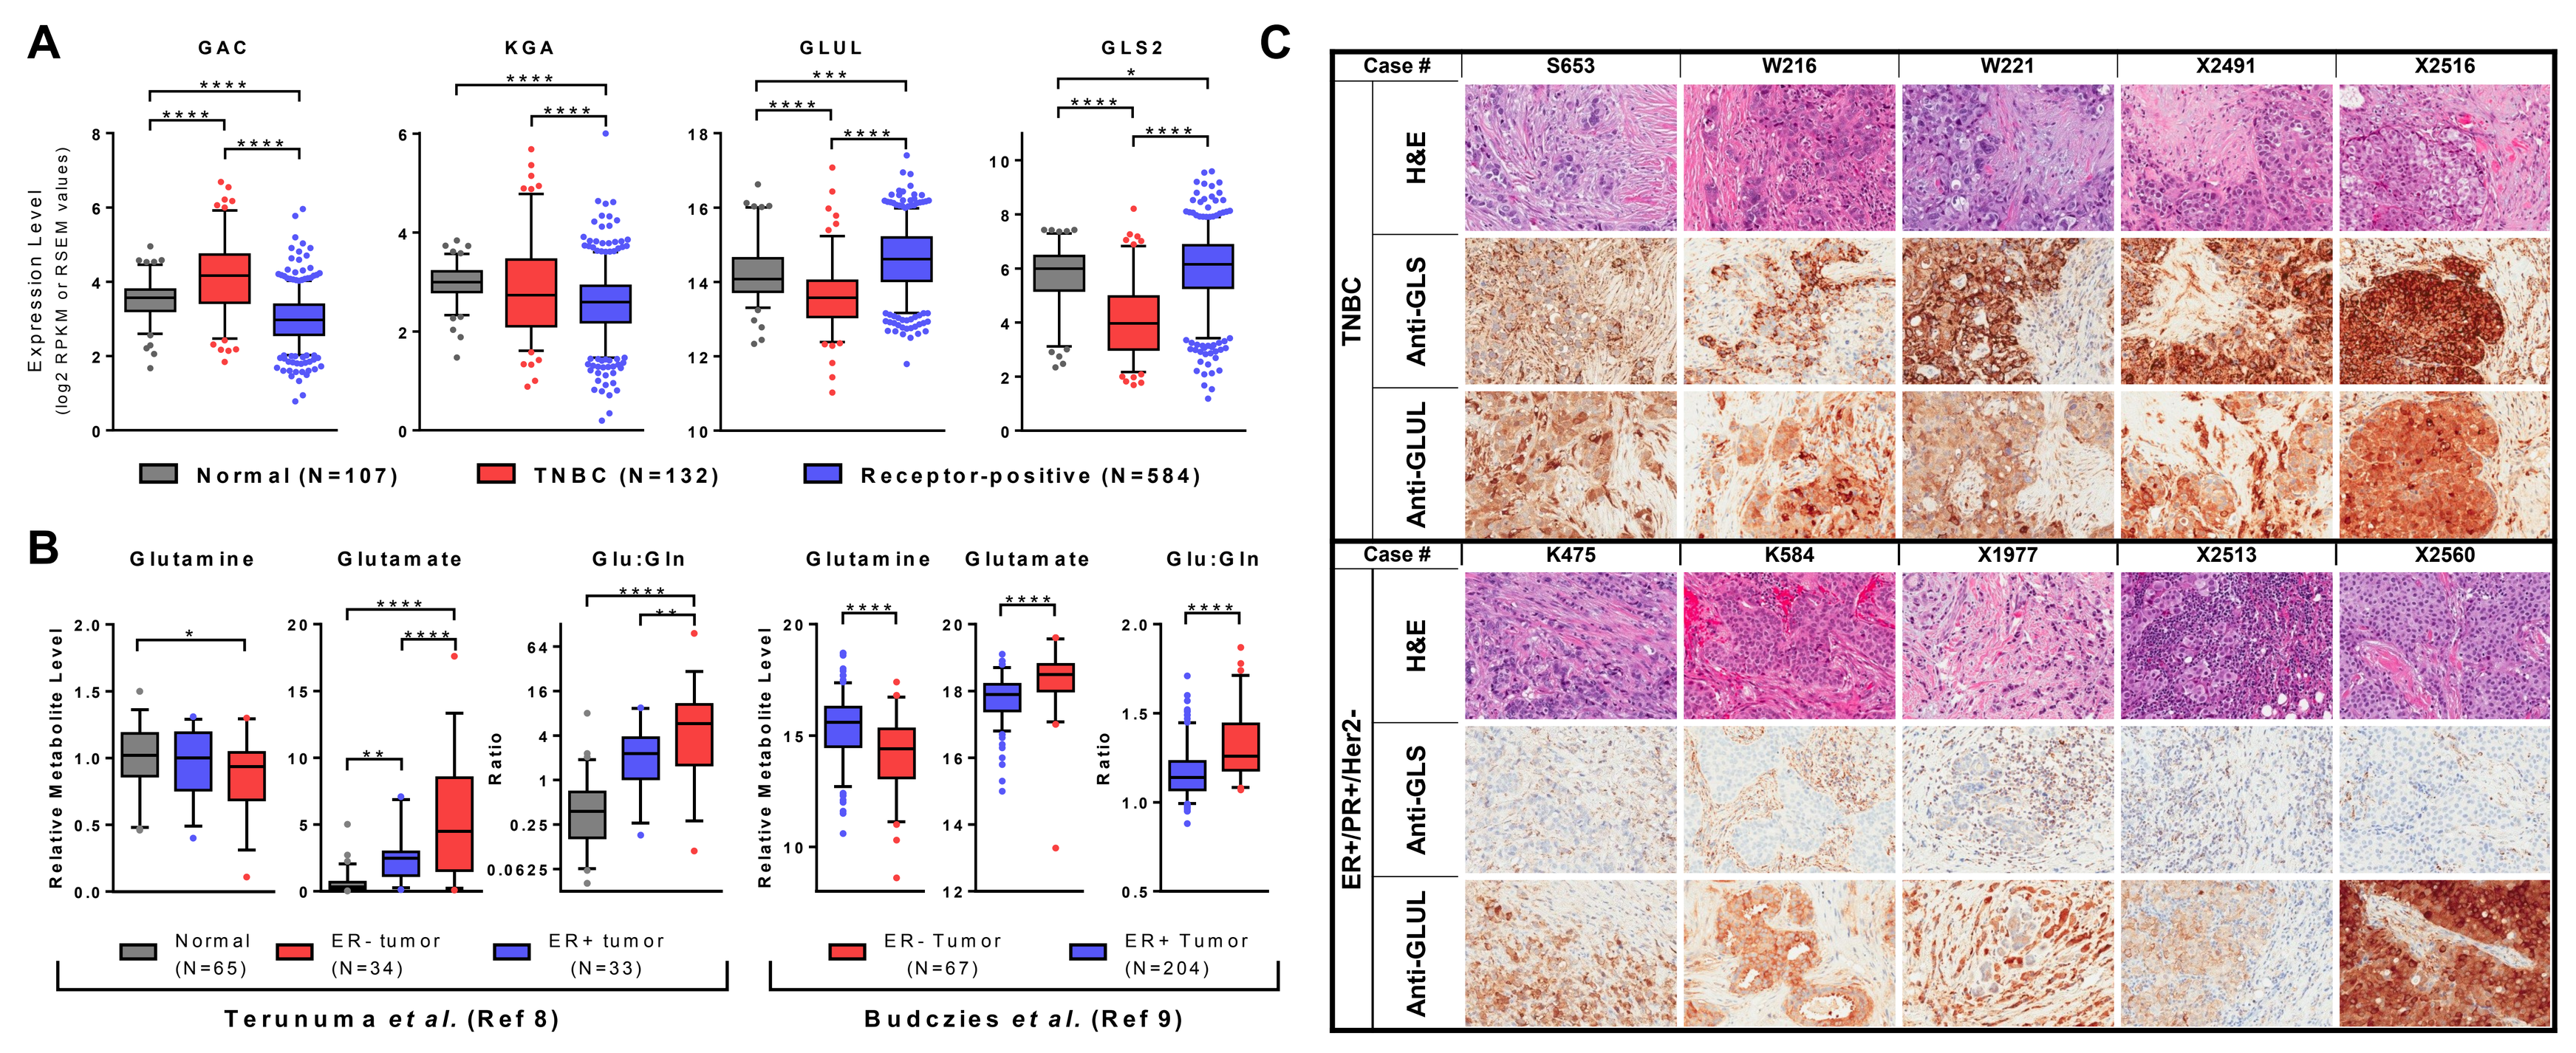

A
GAC
KGA
GLUL
GLS2
Expression Level (log2 RPKM or RSEM values)
Normal (N=107)
TNBC (N=132)
Receptor-positive (N=584)
B
Glutamine
Glutamate
Glu:Gln
Relative Metabolite Level
Ratio
Normal (N=65)
ER- tumor (N=34)
ER+ tumor (N=33)
Terunuma et al. (Ref 8)
ER- Tumor (N=67)
ER+ Tumor (N=204)
Budczies et al. (Ref 9)
C
Case #
S653
W216
W221
X2491
X2516
TNBC
H&E
Anti-GLS
Anti-GLUL
K475
K584
X1977
X2513
X2560
ER+/PR+/Her2-

Figure 3. TNBC primary tumors have elevated glutaminase (*GAC*) mRNA and glutaminase protein levels and increased Glu:Gln ratios. ***A*, GAC mRNA levels are elevated in TNBC.** Expression levels for *GAC, KGA, GLUL and GLS2* in normal breast tissue, TNBC tumors, and receptor-positive tumors from the TCGA breast invasive carcinoma dataset. The log2 transformed expression levels for each gene are plotted. ***B*, Glutamate to glutamine ratios are elevated in primary TNBC tumors.** Relative amounts in glutamine and glutamate and Glu:Gln ratios in normal human breast tissue, TNBC samples and ER+ tumor samples from patient samples in published reports (Ref 8 and Ref 9). Data have been re-plotted here for ease of comprehension. *A-B*, "whiskers" span the 5th to 95th percentile with data outside this range shown as individual data points. Statistical comparisons were performed by (A) one-way ANOVA or (B) unpaired t-test to generate P values: *$P \leq 0.05$,**$P \leq 0.01$, ***$P \leq 0.001$, **** $P \leq 0.0001$. **C, GLS protein levels are increased in primary TNBC samples.** Human formalin-fixed paraffin-embedded tumor samples from TNBC patients (upper rows) and ER+ patients (lower rows) were stained with H&E or anti-GLS antibody (Abcam) or anti-GLUL antibody (Millipore). Samples were purchased from Individumed (Hamburg, Germany).

GAC Expression and Glutamate Levels are Elevated in Several Cancer Types

Elevated GAC expression in TNBC, colorectal cancer, lung cancer, lymphoma, multiple myeloma and lymphoma.

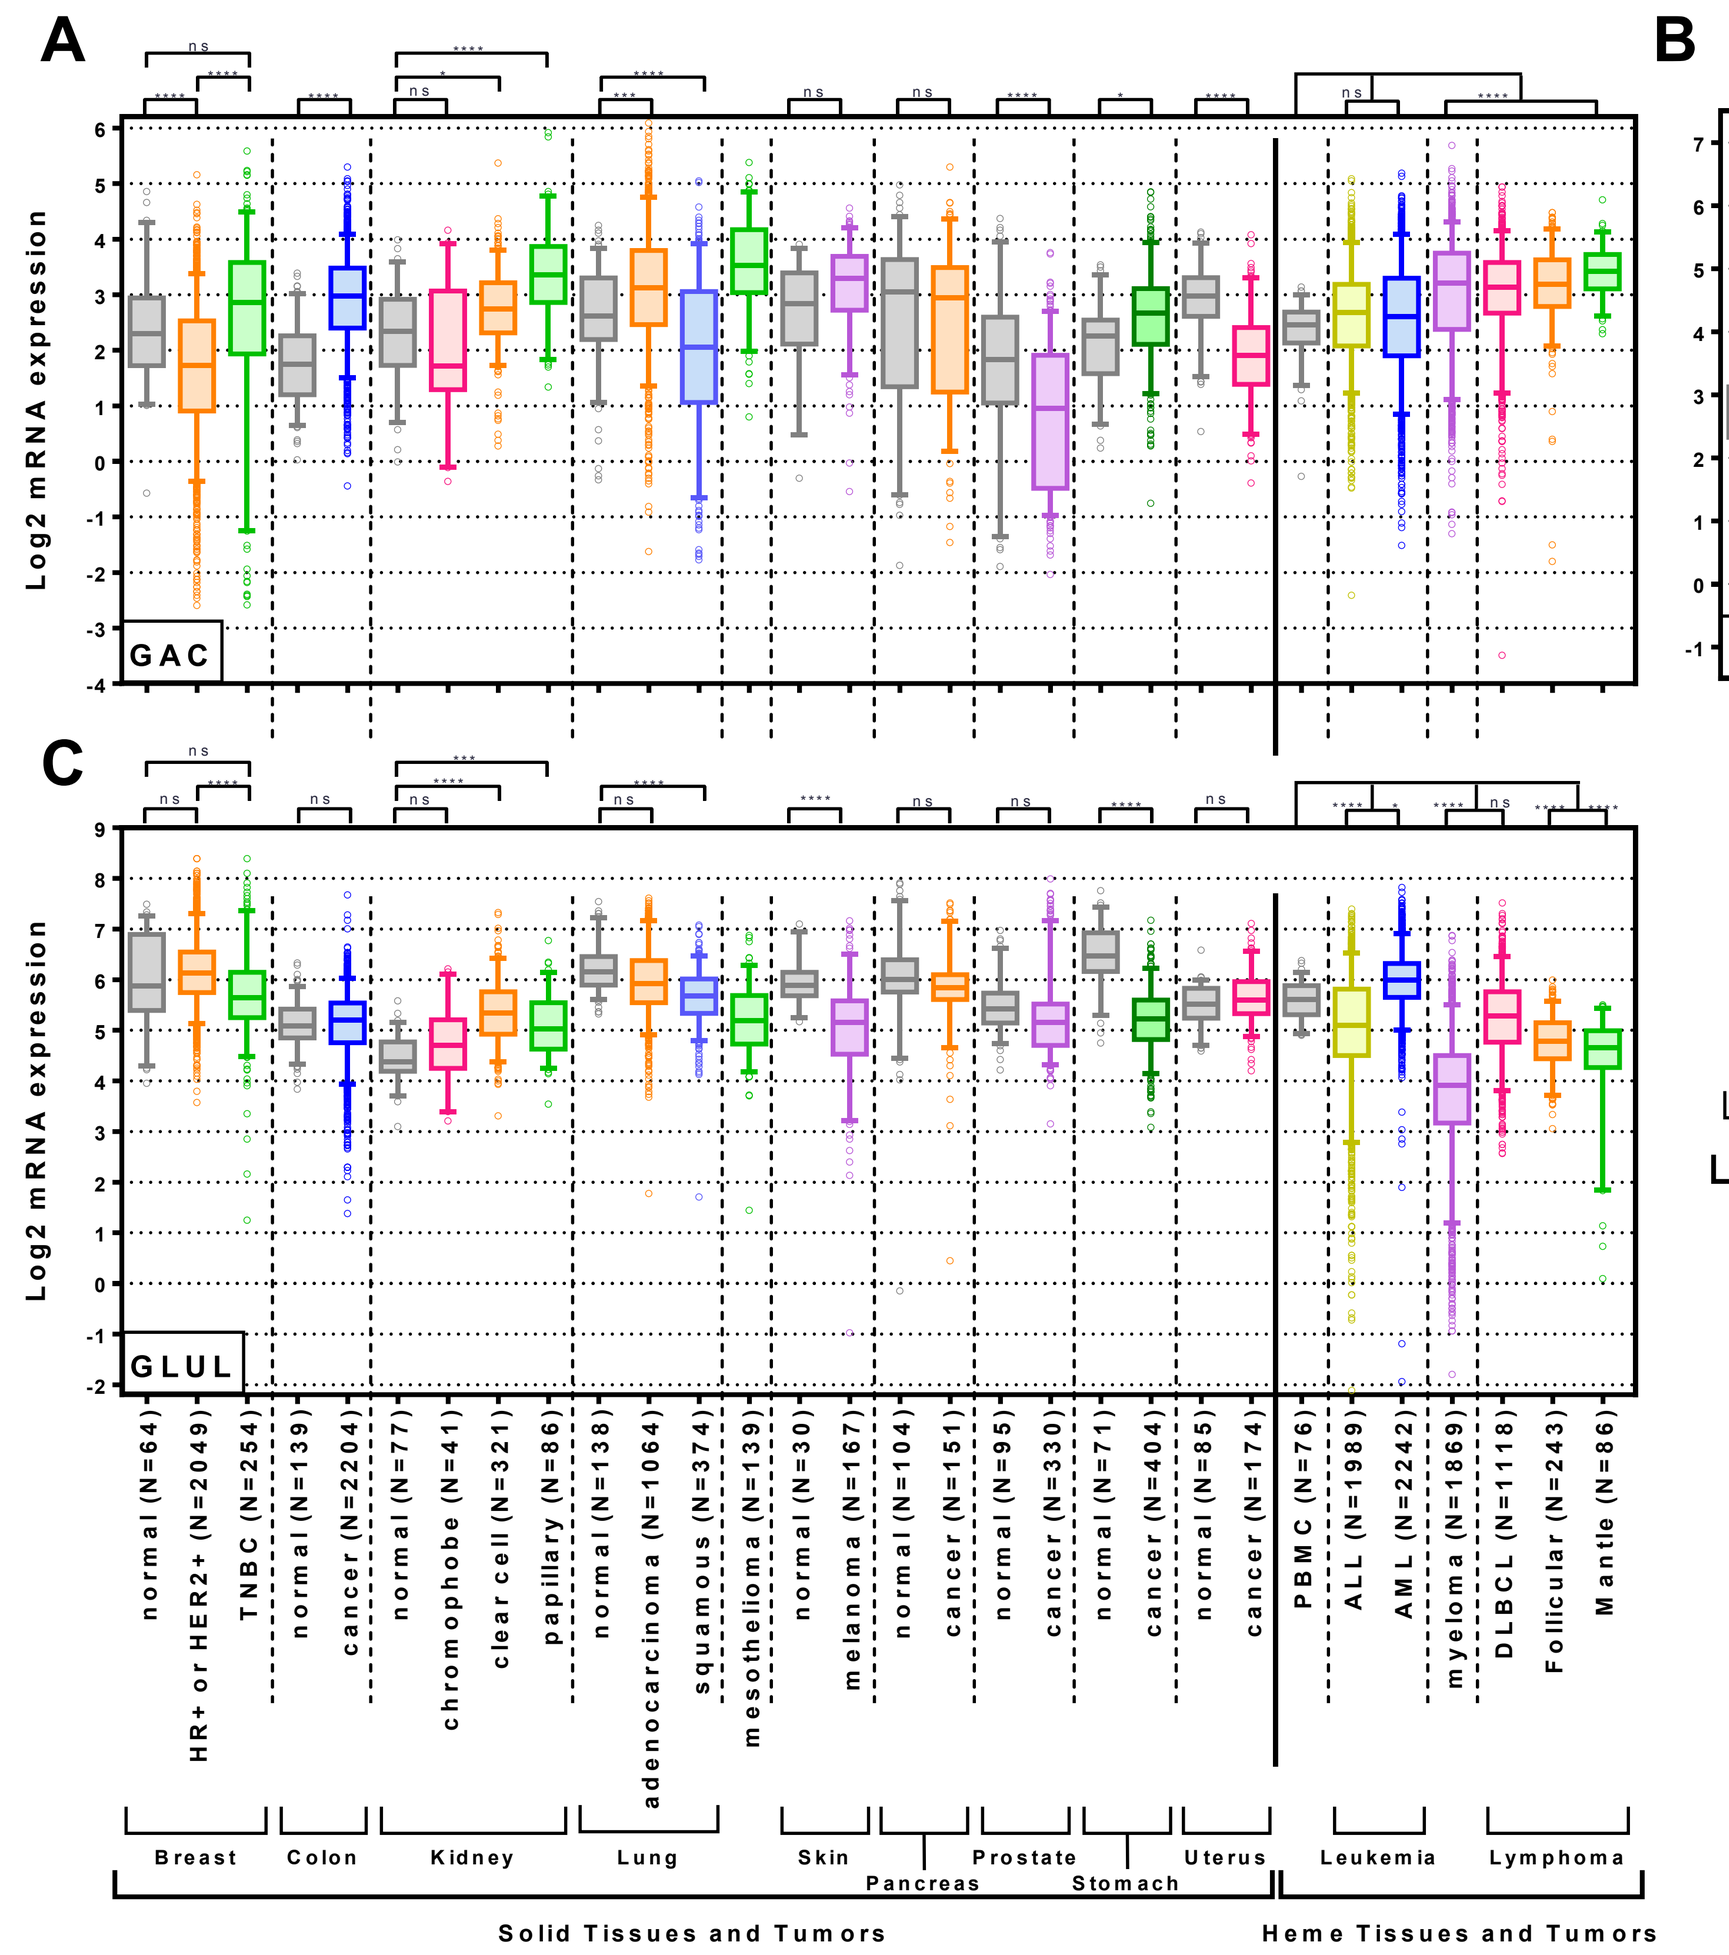

A
C
Log2 mRNA expression
GAC
GLUL
Breast
Colon
Kidney
Lung
Skin
Pancreas
Prostate
Stomach
Uterus
Leukemia
Lymphoma
Solid Tissues and Tumors
Heme Tissues and Tumors

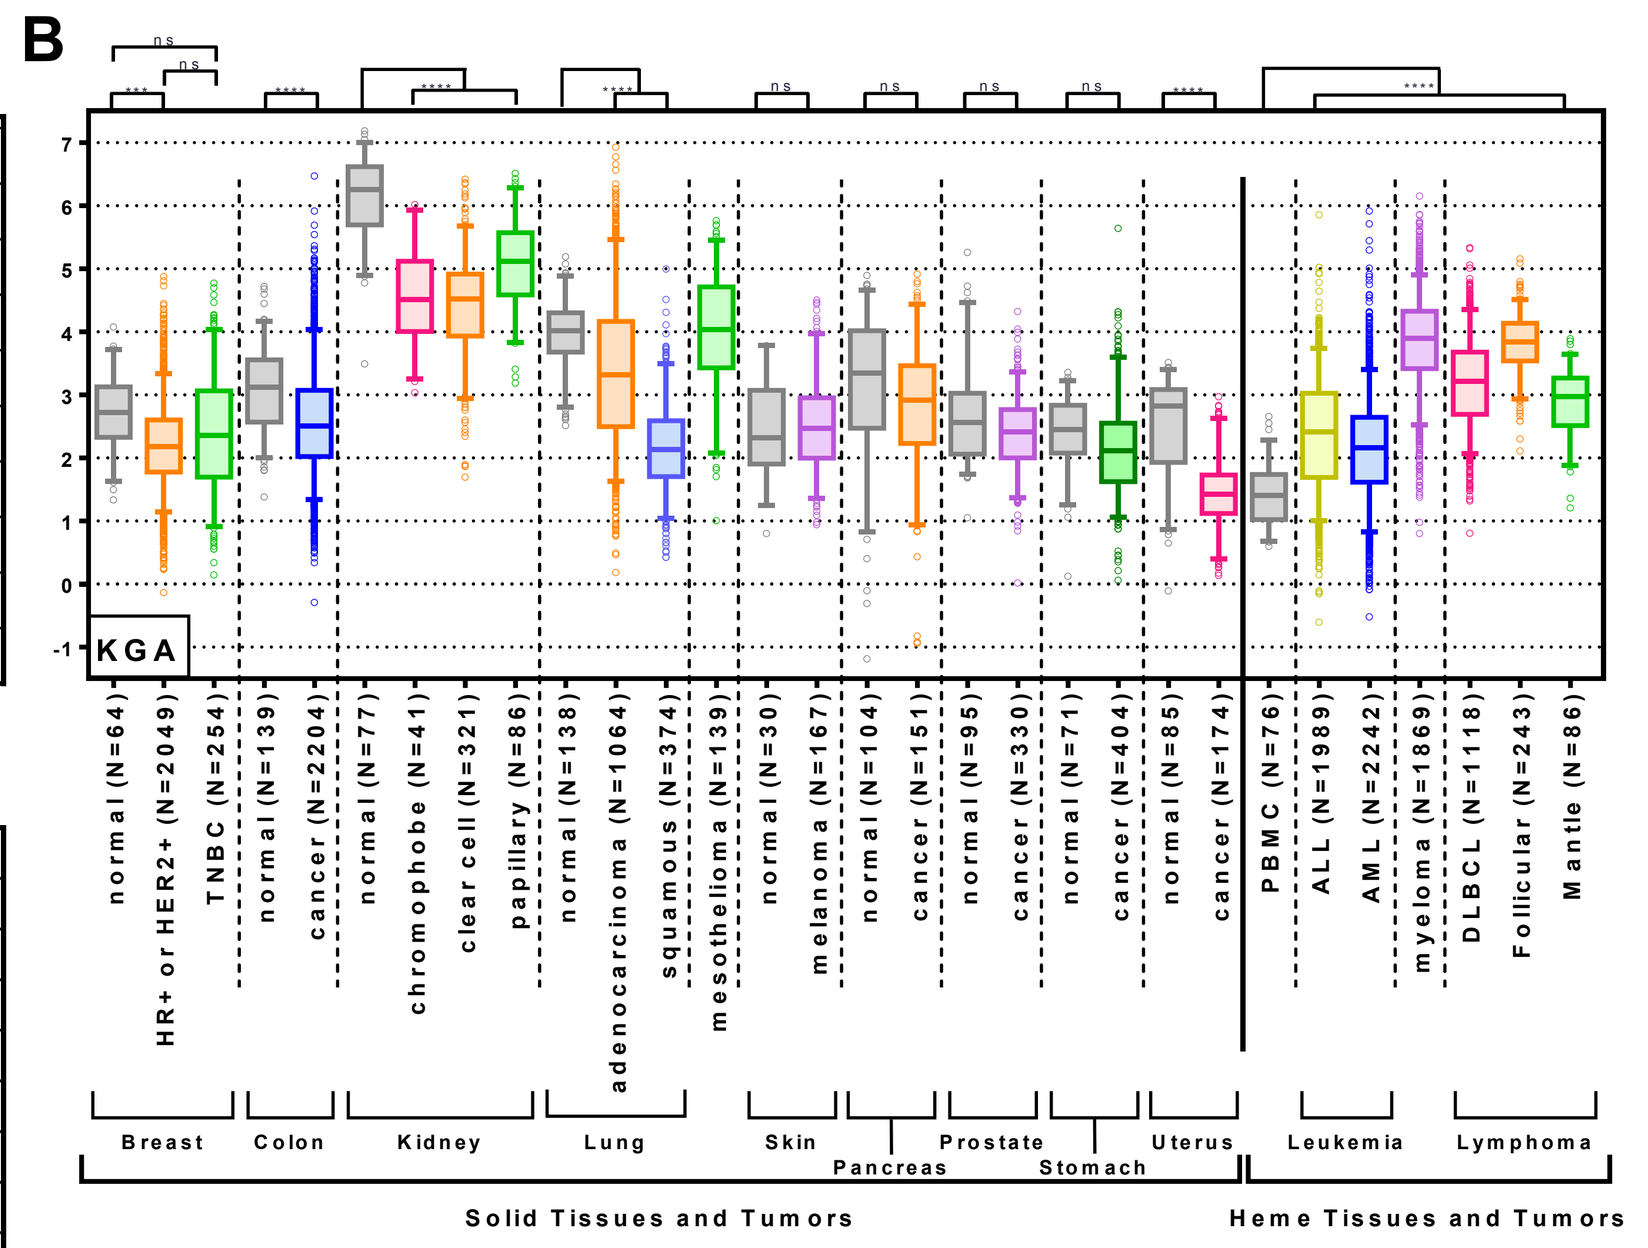

B
KGA
Breast
Colon
Kidney
Lung
Skin
Pancreas
Prostate
Stomach
Uterus
Leukemia
Lymphoma
Solid Tissues and Tumors
Heme Tissues and Tumors

Figure 4: Expression levels of GAC, KGA and GLUL in various cancer types and associated normal tissues. *A*, GAC levels are elevated in triple-negative breast, colon, kidney, lung and stomach cancers as well as multiple myeloma and lymphoma compared to normal tissue. *B*, KGA levels are elevated in leukemia, multiple myeloma and lymphoma compared to normal tissue. C, GLUL levels are reduced in melanoma, stomach cancer and myeloma compared to normal tissue mRNA levels were obtained from Compendia Bioscience™ Translational Bioinformatics Services (Life Technologies, Ann Arbor, MI). mRNA expression are plotted as the log2 RMA normalized values. Whiskers" span the 5th to 95th percentile with data outside this range shown as individual data points. Statistics were performed using One-Way ANOVA to generate a P values: *$P \leq 0.05$,**$P \leq 0.01$, ***$P \leq 0.001$, **** $P \leq 0.0001$, ns (not significant).

Glutamate levels are higher in lung, stomach and colon cancer

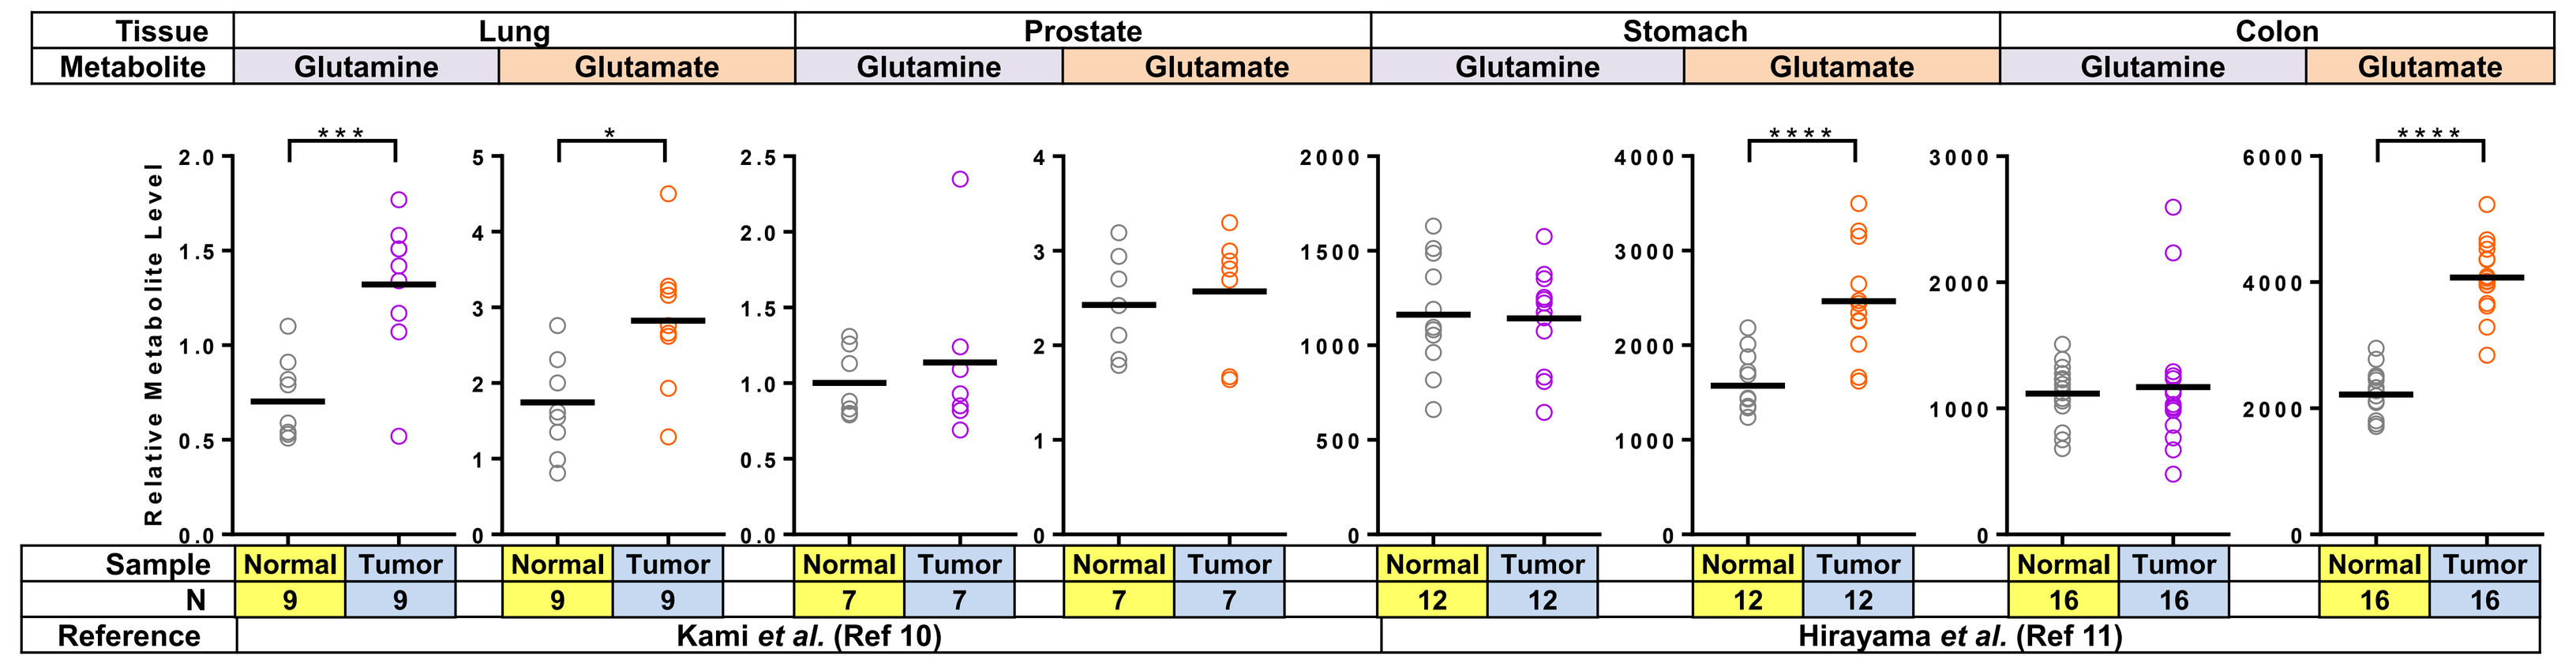

Tissue
Lung
Prostate
Stomach
Colon
Metabolite
Glutamine
Glutamate
Relative Metabolite Level
Sample
Normal
Tumor
N
9
9
9
9
7
7
7
7
12
12
12
12
16
16
16
16
Reference
Kami et al. (Ref 10)
Hirayama et al. (Ref 11)

Figure 5: Glutamate levels are elevated in lung, stomach and colon cancer patients. Primary patient tumor samples from lung, prostate, stomach and colon cancer were analyzed for levels of glutamate and glutamine and published in the indicated references. Data is re-plotted here for ease of comprehension. An unpaired t-test was performed comparing tumor and normal samples to generate P values: *$P \leq 0.05$,**$P \leq 0.01$, ***$P \leq 0.001$, **** $P \leq 0.0001$

Biomarker Analysis in PDX models

Glutamine Utilization Markers are Retained in Primary Tumorgrafts (PDX)

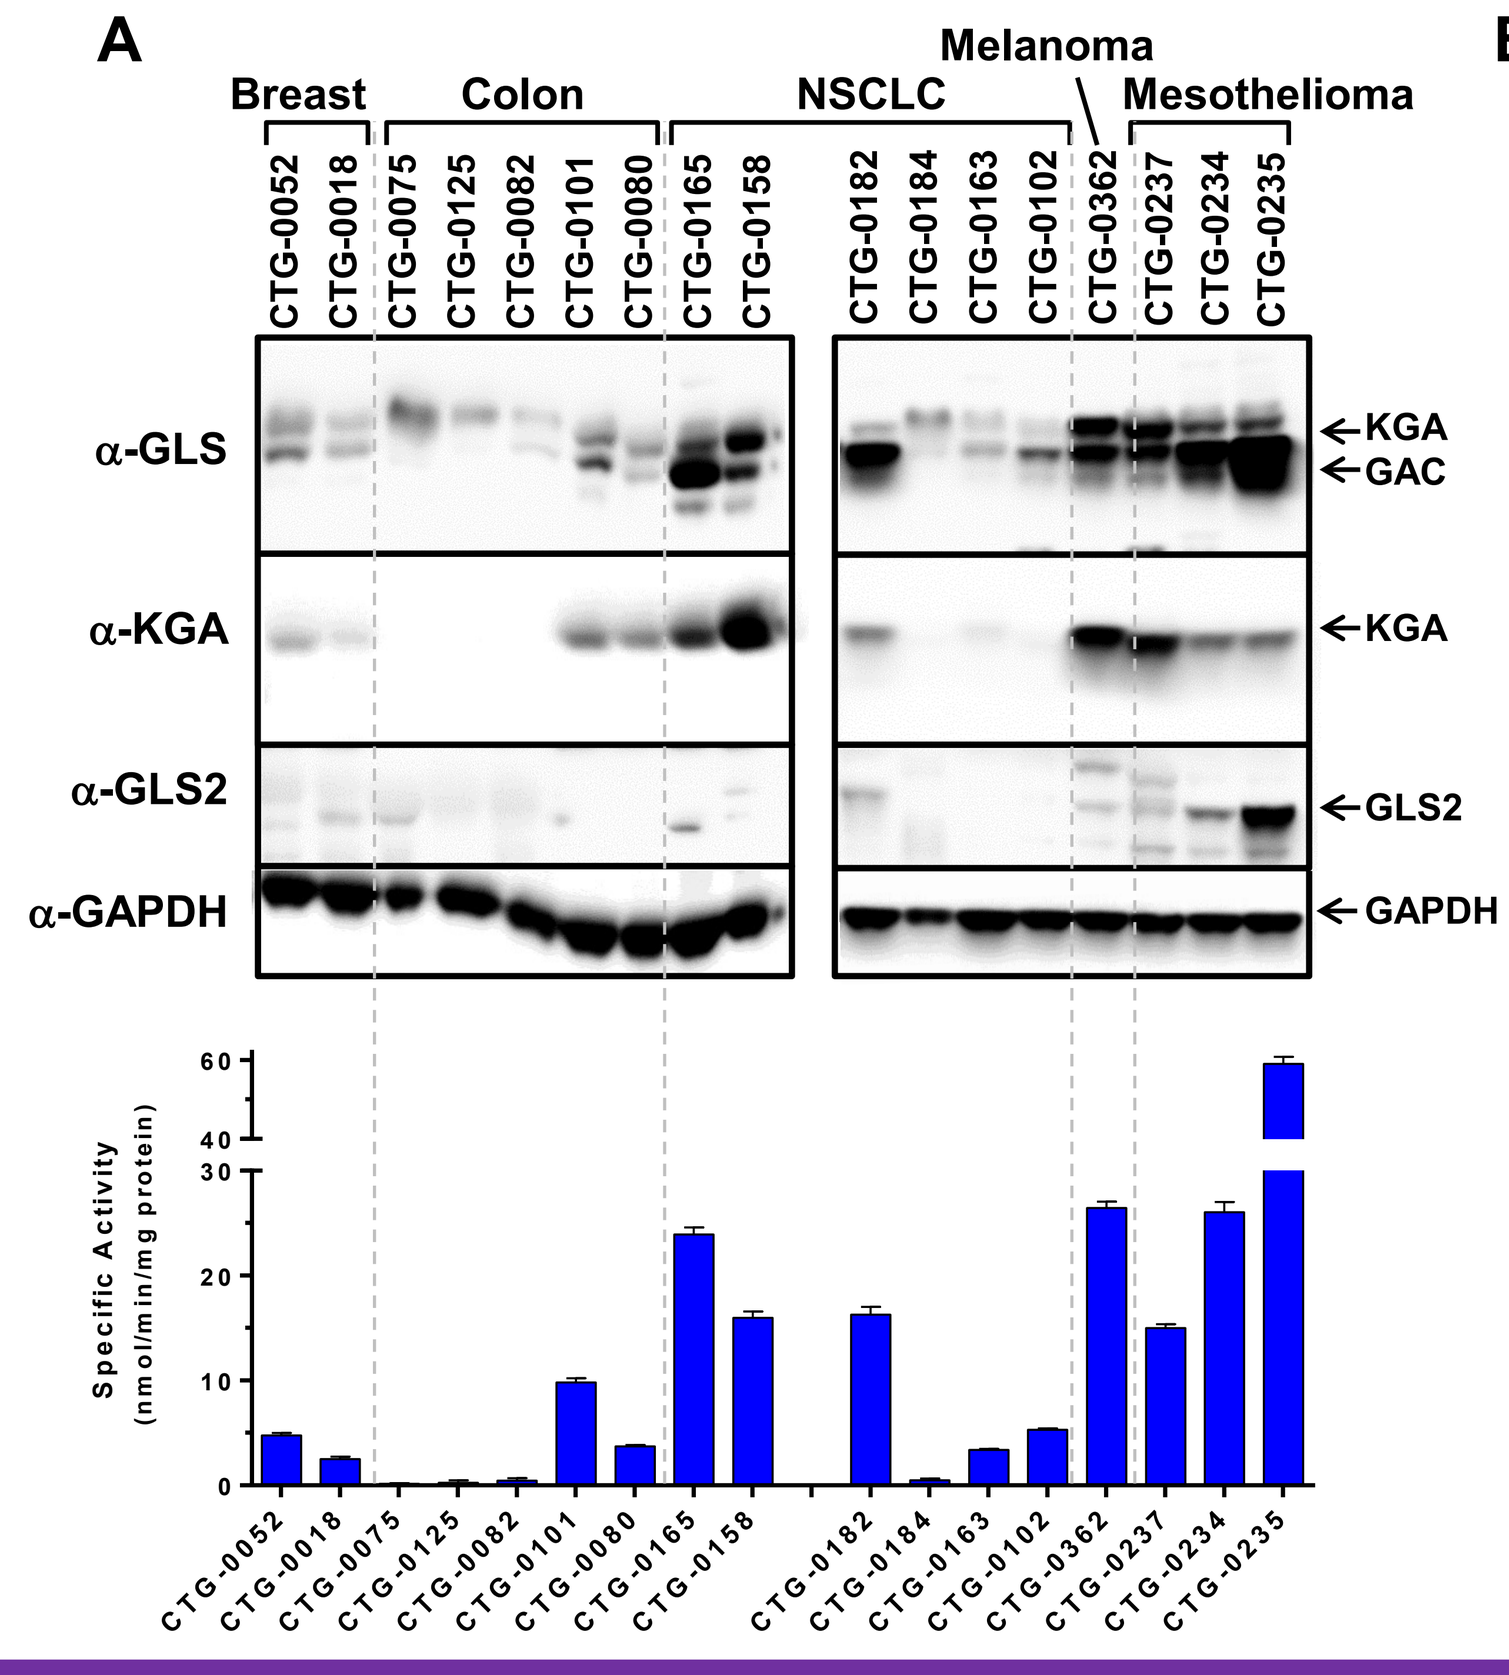

A
Breast
Colon
NSCLC
Melanoma
Mesothelioma
CTG-0052
CTG-0018
CTG-0075
CTG-0125
CTG-0082
CTG-0101
CTG-0080
CTG-0165
CTG-0158
CTG-0182
CTG-0184
CTG-0163
CTG-0102
CTG-0362
CTG-0237
CTG-0234
CTG-0235
α-GLS
α-KGA
α-GLS2
α-GAPDH
KGA
GAC
GLS2
GAPDH
Specific Activity (nmol/min/mg protein)

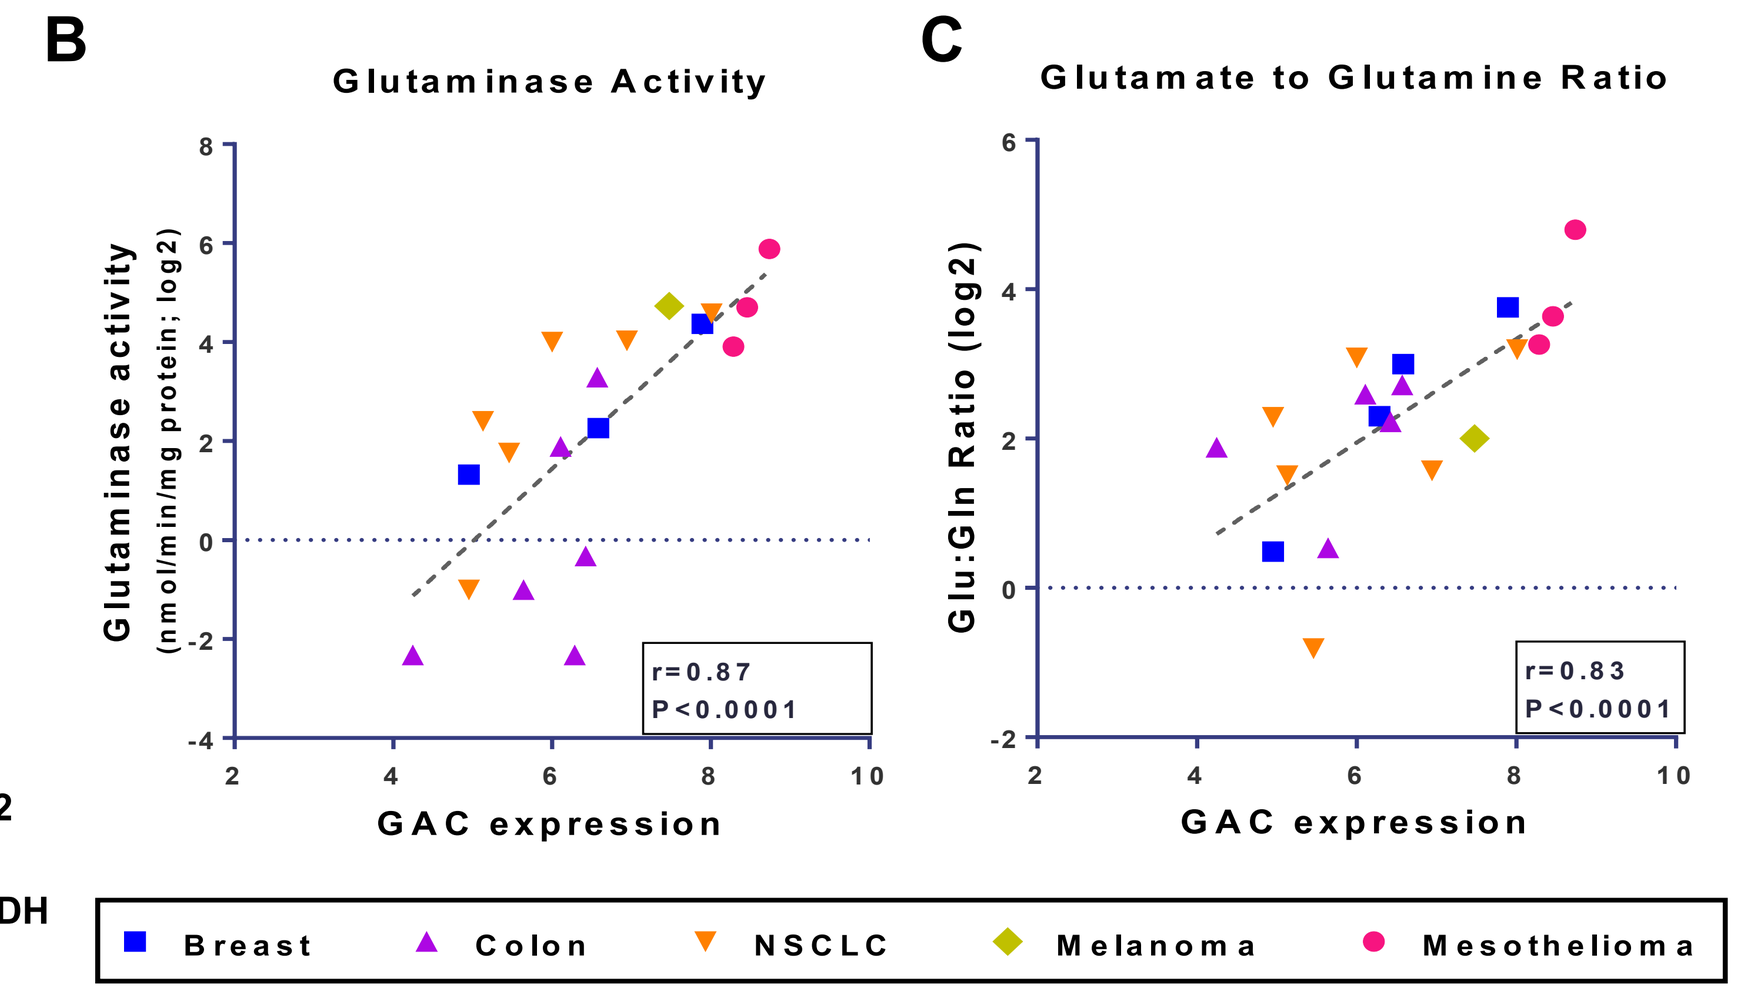

B
Glutaminase Activity
Glutaminase activity (nmol/min/mg protein; log2)
r=0.87
P<0.0001
GAC expression
C
Glutamate to Glutamine Ratio
Glu:Gln Ratio (log2)
r=0.83
P<0.0001
GAC expression
Breast
Colon
NSCLC
Melanoma
Mesothelioma

Figure 6: Glutaminase activity and Glu:Gln ratio correlate with GAC mRNA expression in primary tumorgrafts. *A*, Relationship between GLS specific activity and GLS protein levels. Primary human tumorgrafts were procured from Champions Oncology (Baltimore, MD) and homogenates (20 μg) were prepared and resolved by SDS-PAGE, transferred to nitrocellulose and immunoblotted with the indicated antibodies. The bottom graph shows the corresponding glutaminase specific activity of each tumorgraft homogenate ***B*, Glutaminase activity correlates with GAC mRNA expression.** Correlation between GAC mRNA expression plotted on the x-axis and glutaminase activity plotted on the y-axis. ***C*, Glu:Gln ratio correlates with GAC mRNA expression.** Correlation between GAC mRNA expression plotted on the x-axis tumor Glu:Gln ratio plotted on the y-axis.

CB-839 has anti-Proliferative Activity in Several Cancer Types

The majority of tumor cells responded to CB-839 treatment

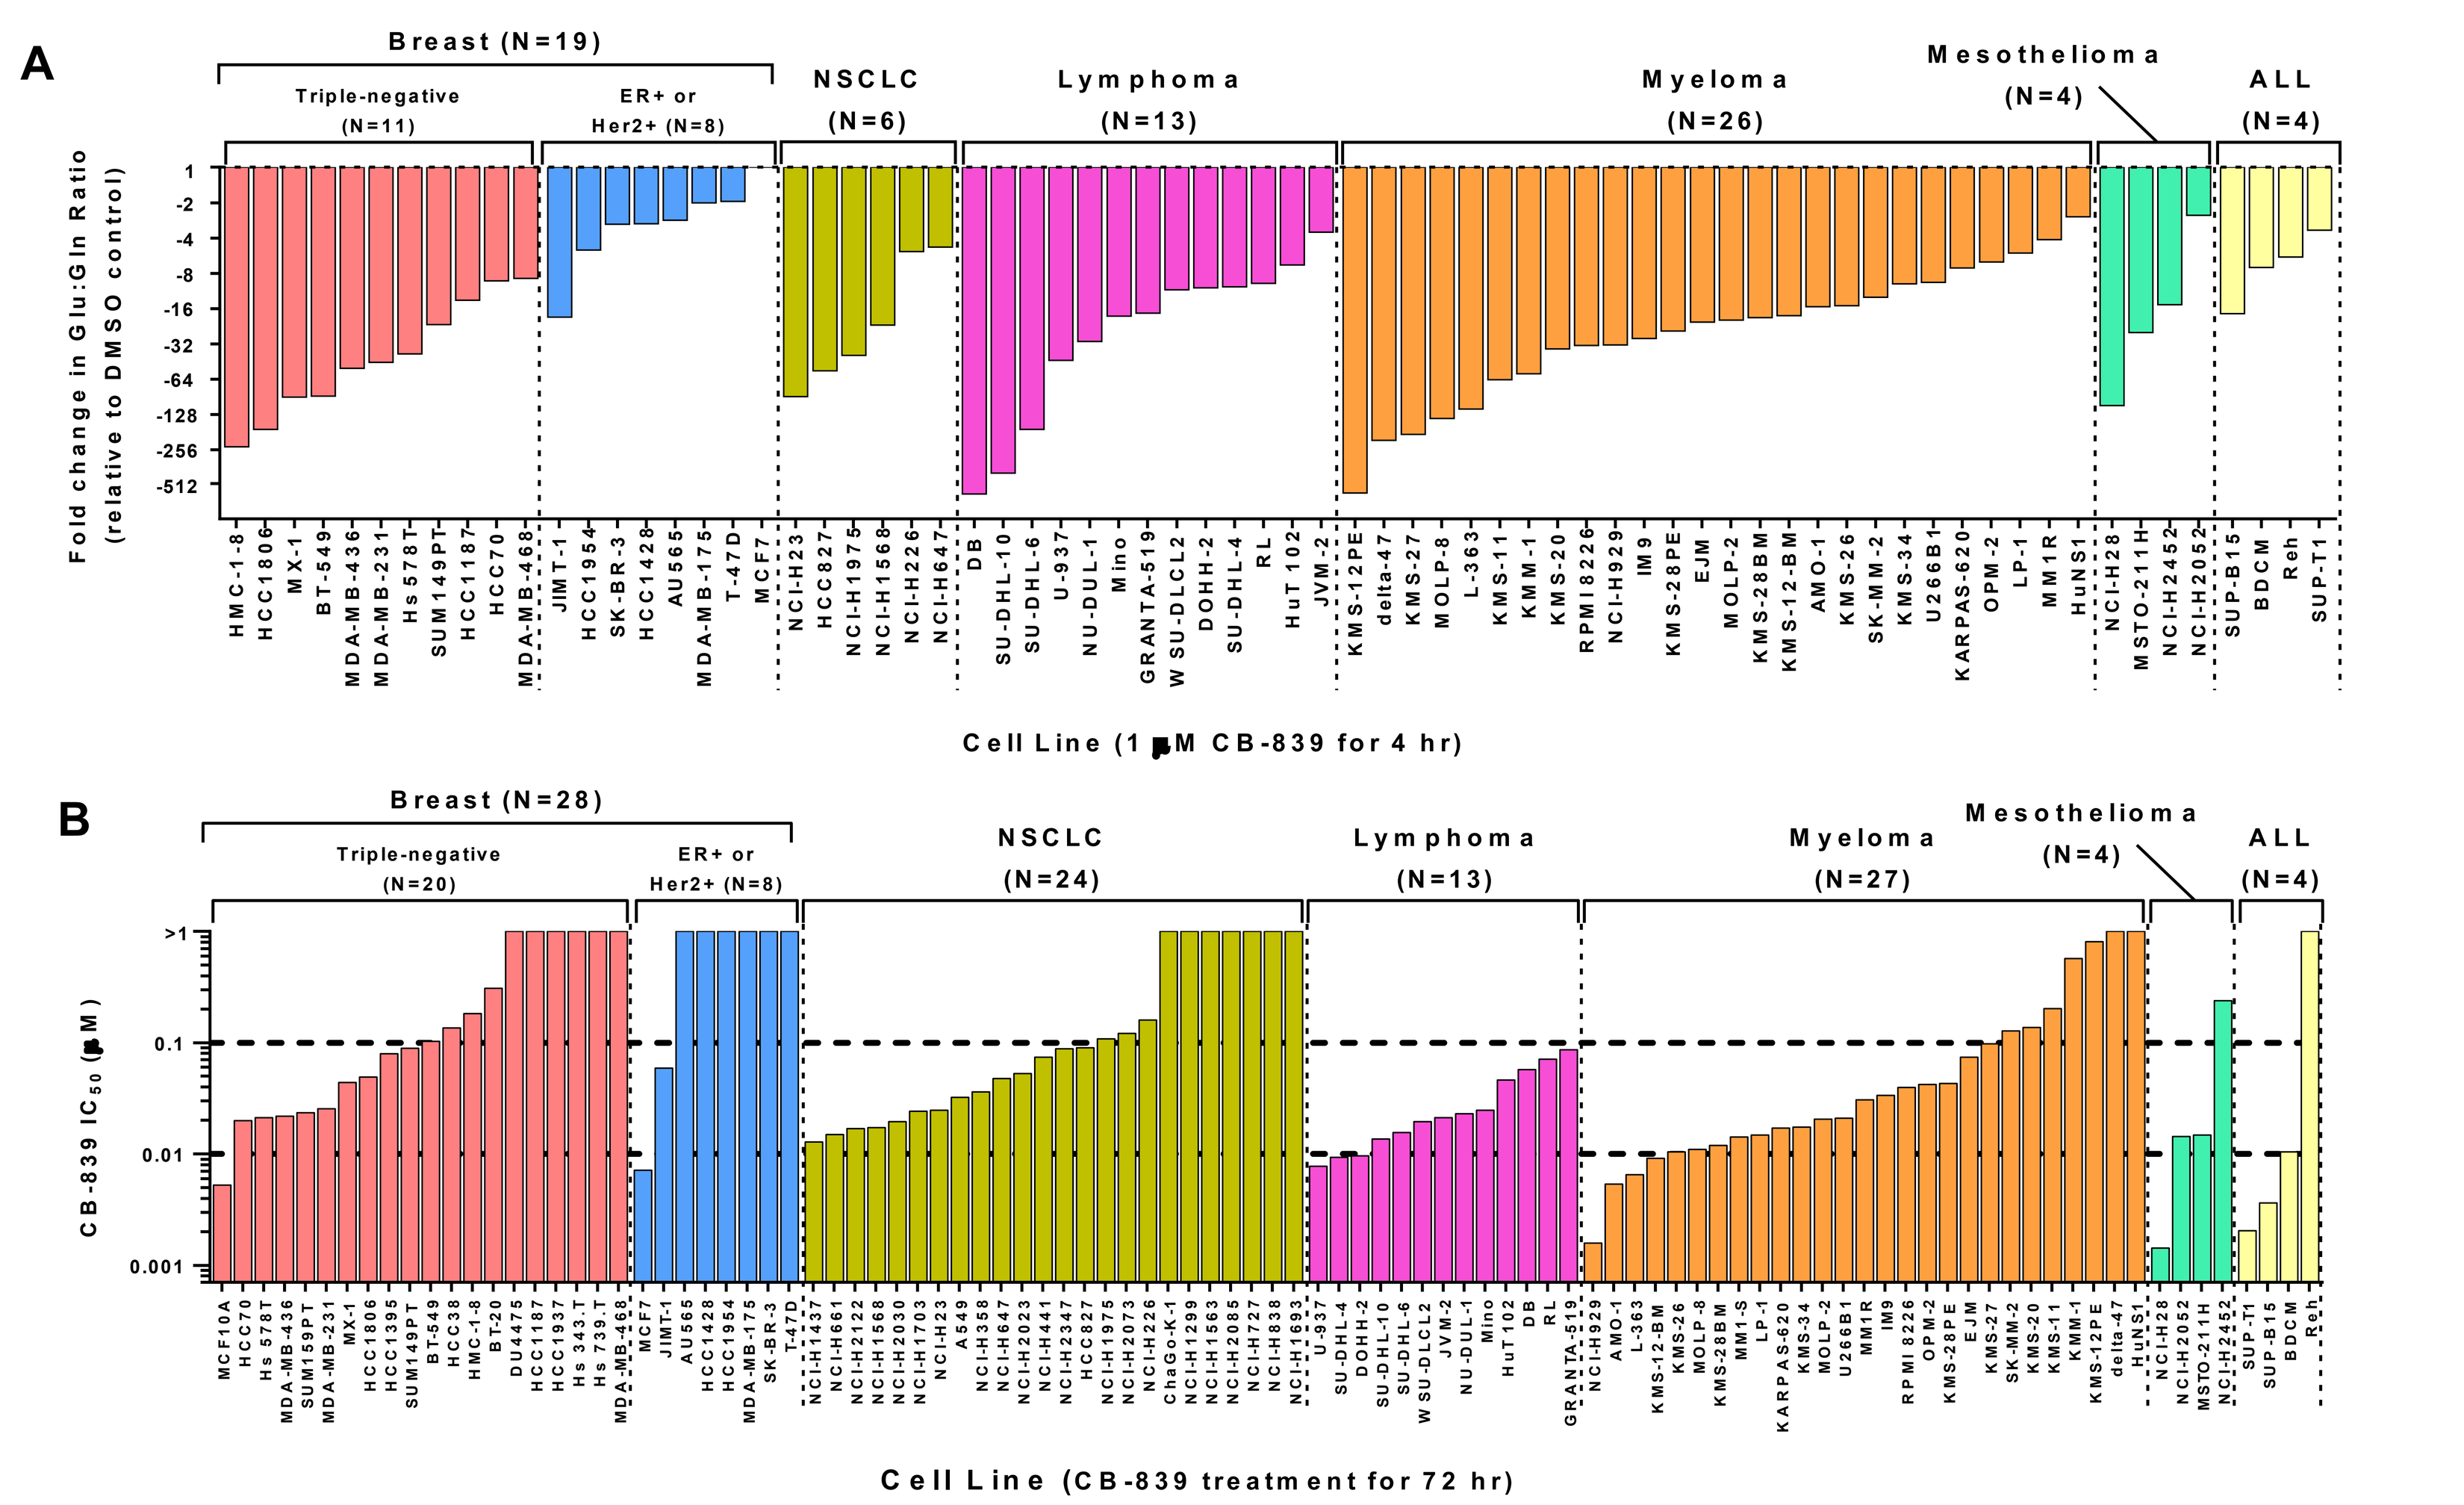

A
Breast (N=19)
Triple-negative (N=11)
ER+ or Her2+ (N=8)
NSCLC (N=6)
Lymphoma (N=13)
Myeloma (N=26)
Mesothelioma (N=4)
ALL (N=4)
Fold change in Glu:Gln Ratio (relative to DMSO control)
Cell Line (1 μM CB-839 for 4 hr)
B
Breast (N=28)
Triple-negative (N=20)
ER+ or Her2+ (N=8)
NSCLC (N=24)
Lymphoma (N=13)
Myeloma (N=27)
Mesothelioma (N=4)
ALL (N=4)
CB-839 IC_{50} (μM)
Cell Line (CB-839 treatment for 72 hr)

Figure 7: CB-839 has anti-proliferative activity in cancer cells representing several histological types. *A*, CB-839 treatment causes an increase in glutamine/glutamate ratio. Cells were treated with 1 μM CB-839 for 4h, harvested and analyzed for levels of glutamine and glutamine by LC/MS/MS. Fold changes in the glutamate to glutamine ratio following CB-839 treatment are plotted for 72 cell lines. ***B*, Treatment with CB-839 has anti-proliferative activity across a panel of tumor cell lines.** Cells were treated with a range of CB-839 concentrations for 72 h and IC_{50} values were determined. The effect on cell proliferation was measured using CellTiterGlo reagent.

The anti-proliferative activity of CB-839 correlates with glutamine withdrawal sensitivity

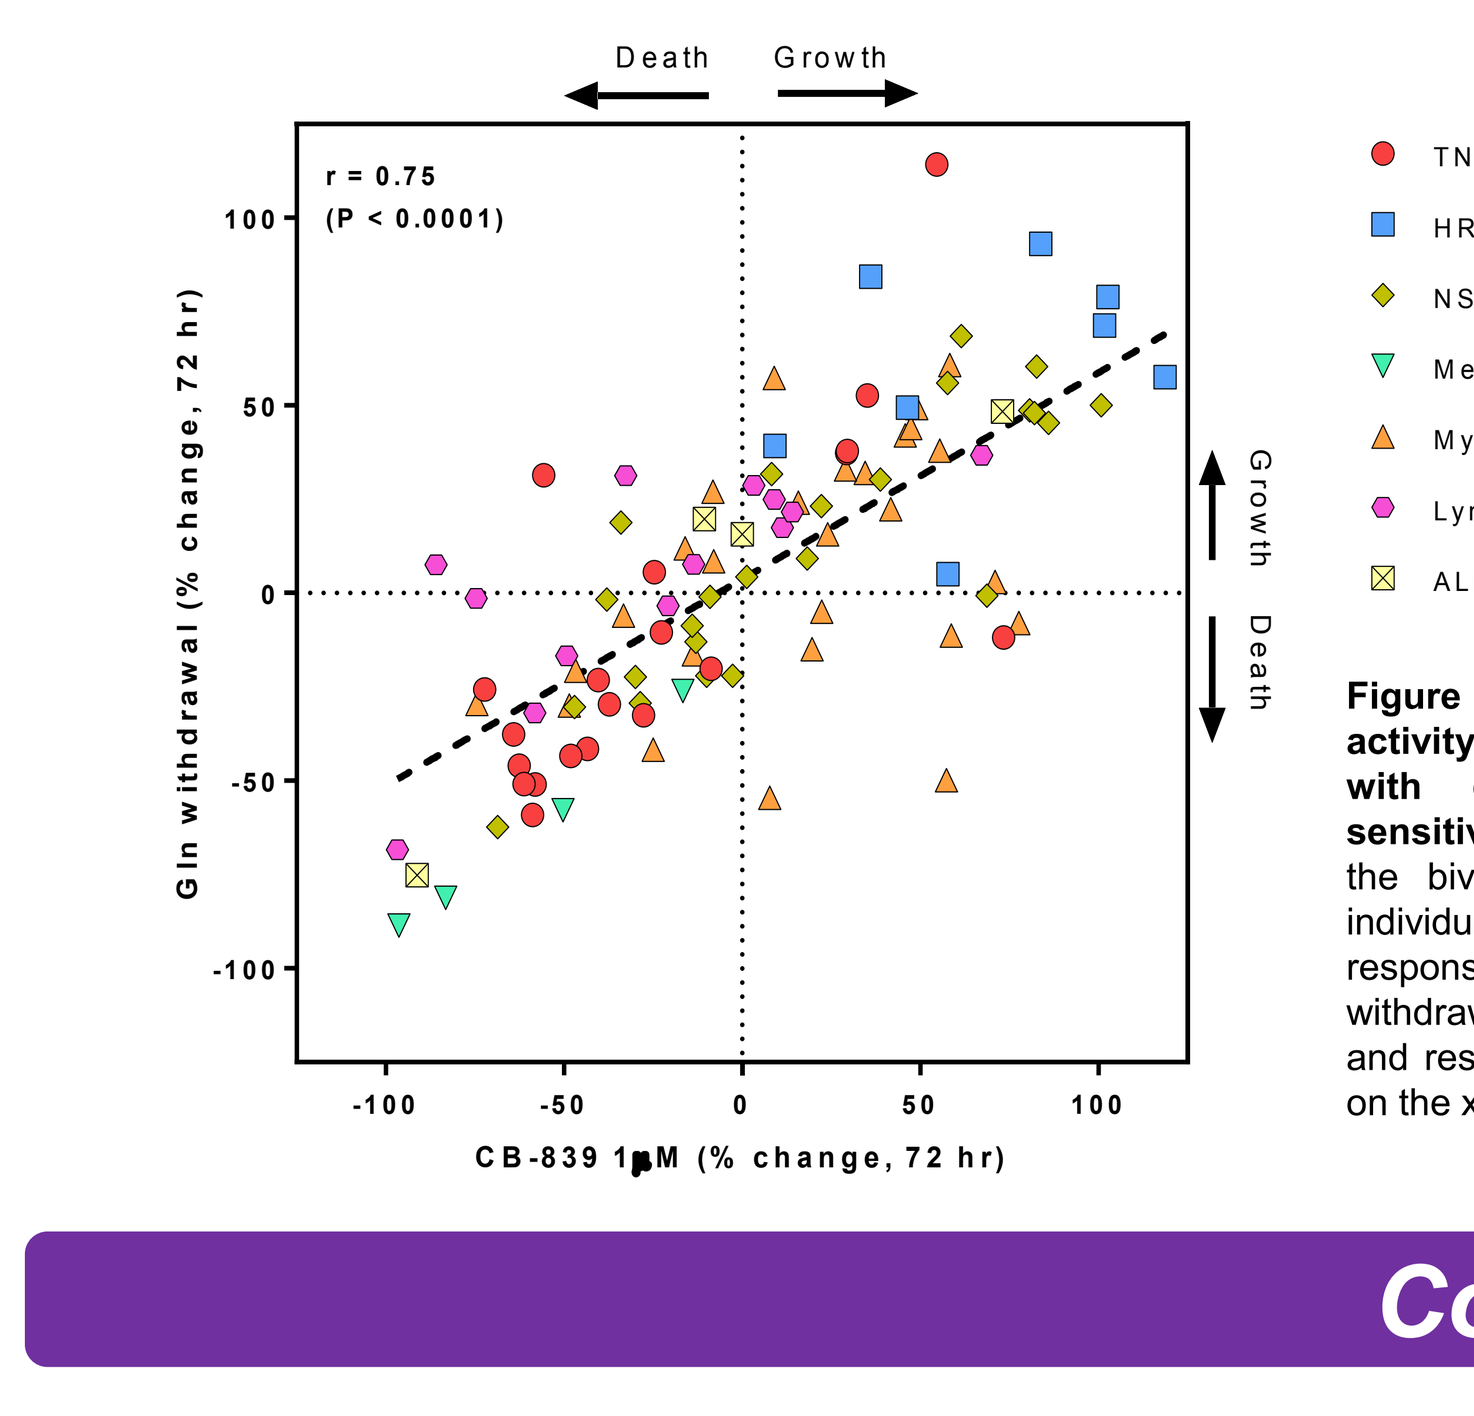

Death
Growth
r = 0.75
(P < 0.0001)
Gln withdrawal (% change, 72 hr)
CB-839 1μM (% change, 72 hr)
TNBC (N=20)
HR+ or HER2+ (N=8)
NSCLC (N=24)
Mesothelioma (N=4)
Myeloma (N=27)
Lymphoma (N=13)
ALL (N=4)

Figure 8: Anti-proliferative activity of CB-839 correlates with glutamine withdrawal sensitivity. Each data point on the bivariate plots depicts an individual cell line with the response to glutamine withdrawal plotted on the y-axis and response to CB-839 plotted on the x-axis.

CB-839 Clinical Trials

The safety, tolerability, pharmacokinetics and pharmacodynamics of CB-839 are being evaluated in the following three ongoing Phase 1 clinical trials:

Clinical Trial*	Patient Population
NCT02071862	All solid tumors during dose escalation; expansion cohorts in TNBC, NSCLC, renal cell carcinoma and mesothelioma at recommended Phase 2 dose
NCT02071888	NHL (B-cell and T-cell), Multiple Myeloma, Waldenstrom's Macroglobulinemia
NCT02071927	Acute Leukemias (AML, ALL)

*** See clinicaltrials.gov for additional details**

Conclusions

- **CB-839 is a potent glutaminase inhibitor with anti-proliferative activity across a panel breast cancer cells that is correlated with: (1) GAC mRNA & protein expression, (2) glutaminase activity, (3) elevated glutamate to glutamine ratio and (4) the triple-negative subtype.**
- **Primary TNBC patient samples show high GAC mRNA expression, GLS protein expression and glutamate to glutamine ratio suggesting that these tumors rely on GLS activity *in vivo*.**
- **Other cancer types display high GAC or KGA mRNA expression (e.g. colon, kidney, lung, mesothelioma, stomach, leukemia, lymphoma, multiple myeloma), high GAC protein expression (lung, colon, mesothelioma, melanoma) and high glutamate levels (lung, colon and stomach) suggesting that these cancers are glutamine avid.**
- **Numerous cell lines representing breast, lung, mesothelioma, multiple myeloma, leukemia, lymphoma show a strong pharmacodynamic response to CB-839. The anti-proliferative response to CB-839 is correlated with sensitivity to glutamine withdrawal suggesting that CB-839 may have anti-tumor activity in these clinical settings.**
- **We have initiated clinical trials in solid and hematological malignancies to test the tolerability, exposure and efficacy of CB-839.**

References

1. Shanware *et al.* (2011) *J Mol Med* 89:229-36
2. Cheng *et al.* (2011) *PNAS* 108:8674-9
3. Gao *et al.* (2009) *Nature* 458, 762-5
4. Le *et al.* (2012) *Cell Metab* 15:110-21
5. van den Heuvel *et al.* (2012) *Cancer Biol Ther* 13:1185-94
6. Cassago *et al.* (2012) PNAS 109:1092-7
7. Gross *et al.* (2014) *Mol Cancer Ther* 13: in press
8. Terunuma *et al.* (2014) *J Clin Invest* 124:398-412
9. Budczies *et al.* (2013) *J Proteomics* 94:279-88
10. Kami *et al.* (2013) *Metabolomics* 9:444-53
11. Hirayama *et al.* (2009) *Cancer Res* 69:4918-25